Identifying T-cell engagers with optimal potency & cytokine release profiles

with differentiated and developable CD3-binding antibodies

AUTHORS
Juntao (Matt) Mai, Kate Caldwell, Lindsay DeVorkin, Grace P. Leung, Karine Herve, Yuri Hwang, Cristina Faralla, Wei Wei, Emma Lathouwers, Valentine de Puyraimond, Lauren Clifford, Rhys S. Chappell, Stefan Hannie, Patrick Farber, Katherine J. Lam, Harveer Dhupar, Tran N. T. Tran, Melissa Cid, Lena M. Bolten, Tova Pinsky, Jessica Fernandes Scortecci, Ping Xiang, Courteney Lai, Ahn Lee, Vivian Z. Li, Patrick Chan, Jasmine Chin, Steve Booth, Amy C. H. Lee, Allison Goodman, Stephanie K. Masterman, Sherie Duncan, Aaron Yamniuk, Kush Dalal, Tim M. Jacobs, Raffi Tonikian, **Bryan C. Barnhart*** *presenter

AUTHORS' AFFILIATION
AbCellera, Vancouver, Canada

DOWNLOAD POSTER

#1886

THE PROBLEM

Most T-cell engagers are built from sub-optimal CD3-binders

CD3 T-cell engagers have the potential to be powerful cancer treatments, but the small number of available CD3-binding antibodies and limited multispecific engineering technologies have been barriers to development.

Approximately 80% of T-cell engagers in clinical trials are engineered with CD3-binding antibodies derived from SP34-2, a mouse antibody discovered in 1985 that binds an unstructured linear epitope on the N-terminus of CD3ε[1]. SP34-2 and other CD3-binders such as OKT3 and UCHT1 are often heavily engineered to reduce the risk of cytokine release syndrome and to improve developability properties.

THE GOAL

Build optimal T-cell engagers from novel CD3-binding antibodies

Identifying T-cell engagers that balance anti-tumor potency with potential toxicities, such as cytokine release syndrome, requires simultaneous tuning of both the CD3- and tumor-binding arms. Pairs of antibodies that achieve this balance are rare, creating a need for diverse and developable antibodies that can be combined and tested at scale to identify optimal clinical candidates.

To streamline T-cell engager development, we discovered and characterized novel CD3-binding antibodies with binding and functional properties that are distinct from commonly used CD3-binders, including SP34-2.

To validate our CD3-binding antibodies, we used our bispecific engineering platform, OrthoMab™, to generate proof-of-concept T-cell engagers targeting either EGFR or PSMA. We then compared tumor cell killing and cytokine release profiles to benchmark-derived molecules across different tumor cell lines.

THE OUTCOME

T-cell engagers with high potency and low cytokine release

We used our high-throughput T-cell engager platform to characterize the resulting bispecific antibodies. The results demonstrate that:

- T-cell engagers engineered from AbCellera's portfolio of differentiated CD3-binding antibodies have functional profiles superior to benchmark molecules
- OrthoMab™-engineered T-cell engagers have favorable developability and PK properties
- The optimal CD3-binder depends on several factors, including tumor target and target density

Together, these studies illustrate that combining novel CD3-binding antibodies, clinically validated multispecific engineering technologies, and an integrated discovery and development engine streamlines identification of optimal T-cell engagers for diverse tumor targets.

T-cell engagers with optimal functional properties

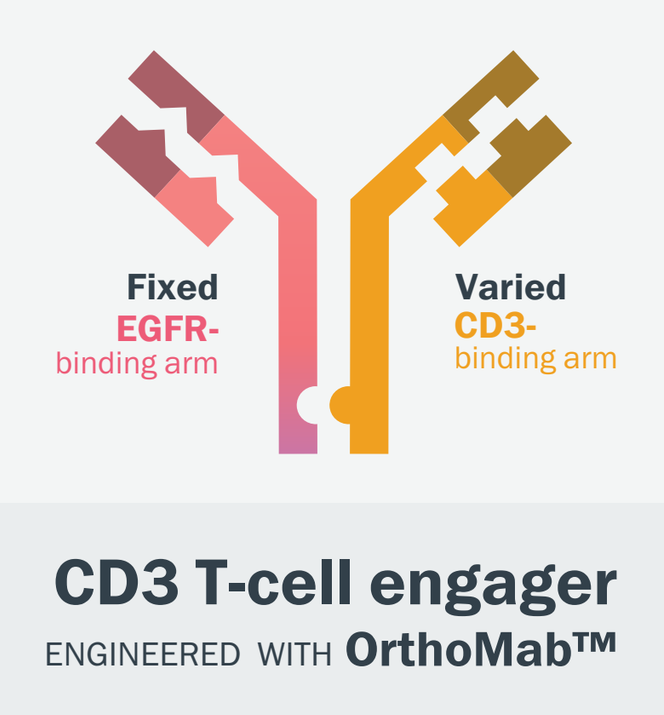
Fixed
EGFR-
binding arm
Varied
CD3-
binding arm
CD3 T-cell engager
ENGINEERED WITH OrthoMab™

We generated proof-of-concept T-cell engagers with high potency and low cytokine release.

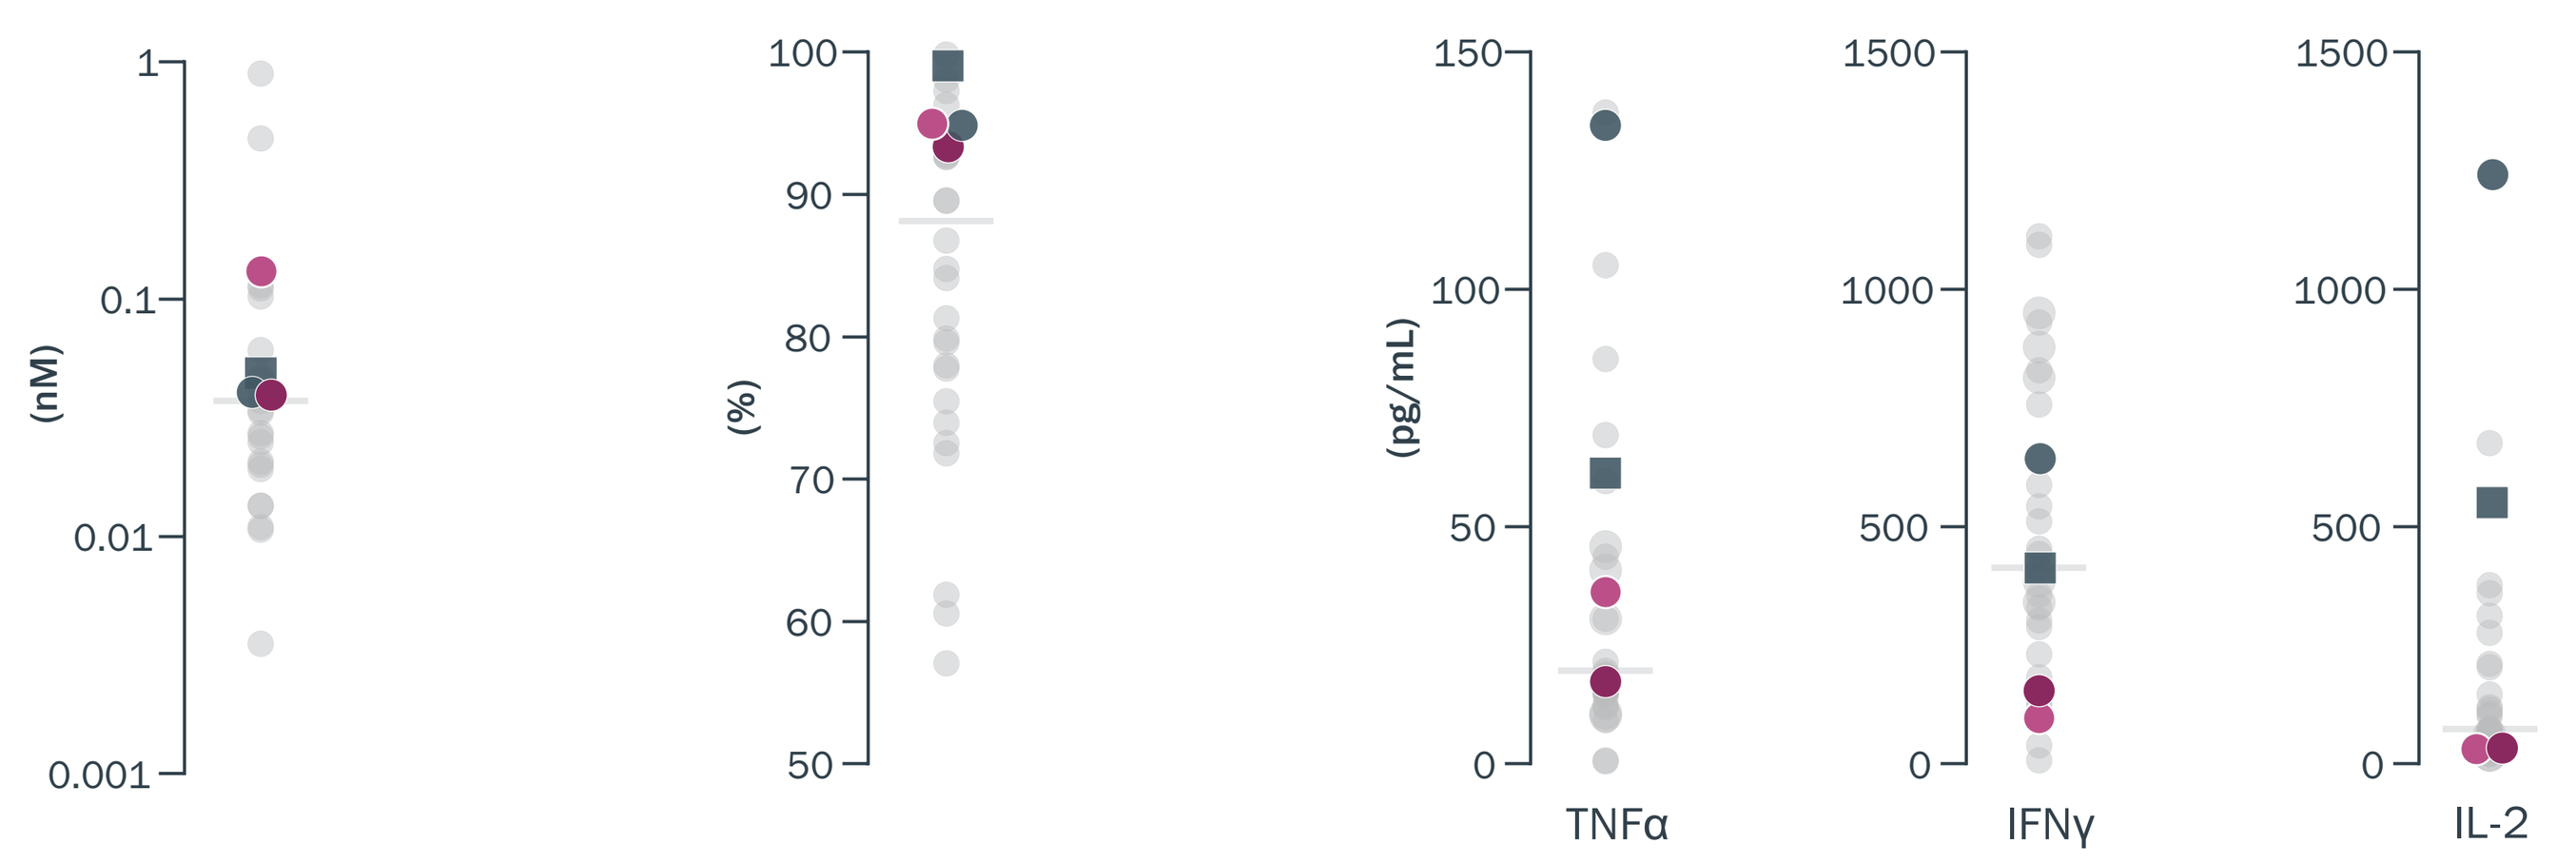
Tumor cell killing
Potency (EC50)
Maximum killing
Cytokine release
Maximum [cytokine]
(nM)
(%)
(pg/mL)
TNFα
IFNγ
IL-2

CD3 arms	Affinity (nM)	
AbCellera-discovered		
CD3-1	εδ (351)	εγ (171)
CD3-2	εγ (258)	
others (15)	εδ (0.99 - 350)	εγ (7.44 - 1260)
SP34-derived		
pasotuxizumab[2]	high affinity	
cibisatamab[3]	low affinity	

Figure 1. T-cell engagers engineered using diverse CD3-binding antibodies have broad tumor cell killing and cytokine release profiles. We generated bispecific antibodies with a fixed EGFR-binding arm (matuzumab) and varied CD3-binding arms (AbCellera-discovered or SP34-derived) using our multispecifics engineering platform, OrthoMab™. T cell-mediated killing of HeLa cells was measured using unactivated human T cells incubated with target cells at a ratio of 10:1. Cell killing and cytokine concentrations were measured 48 hours following addition of the bispecific antibodies. Potency (EC_{50}) is the concentration of each bispecific antibody needed to induce 50% of the maximal T cell-mediated killing of HeLa cells.

We identified T-cell engagers with optimal functional profiles.

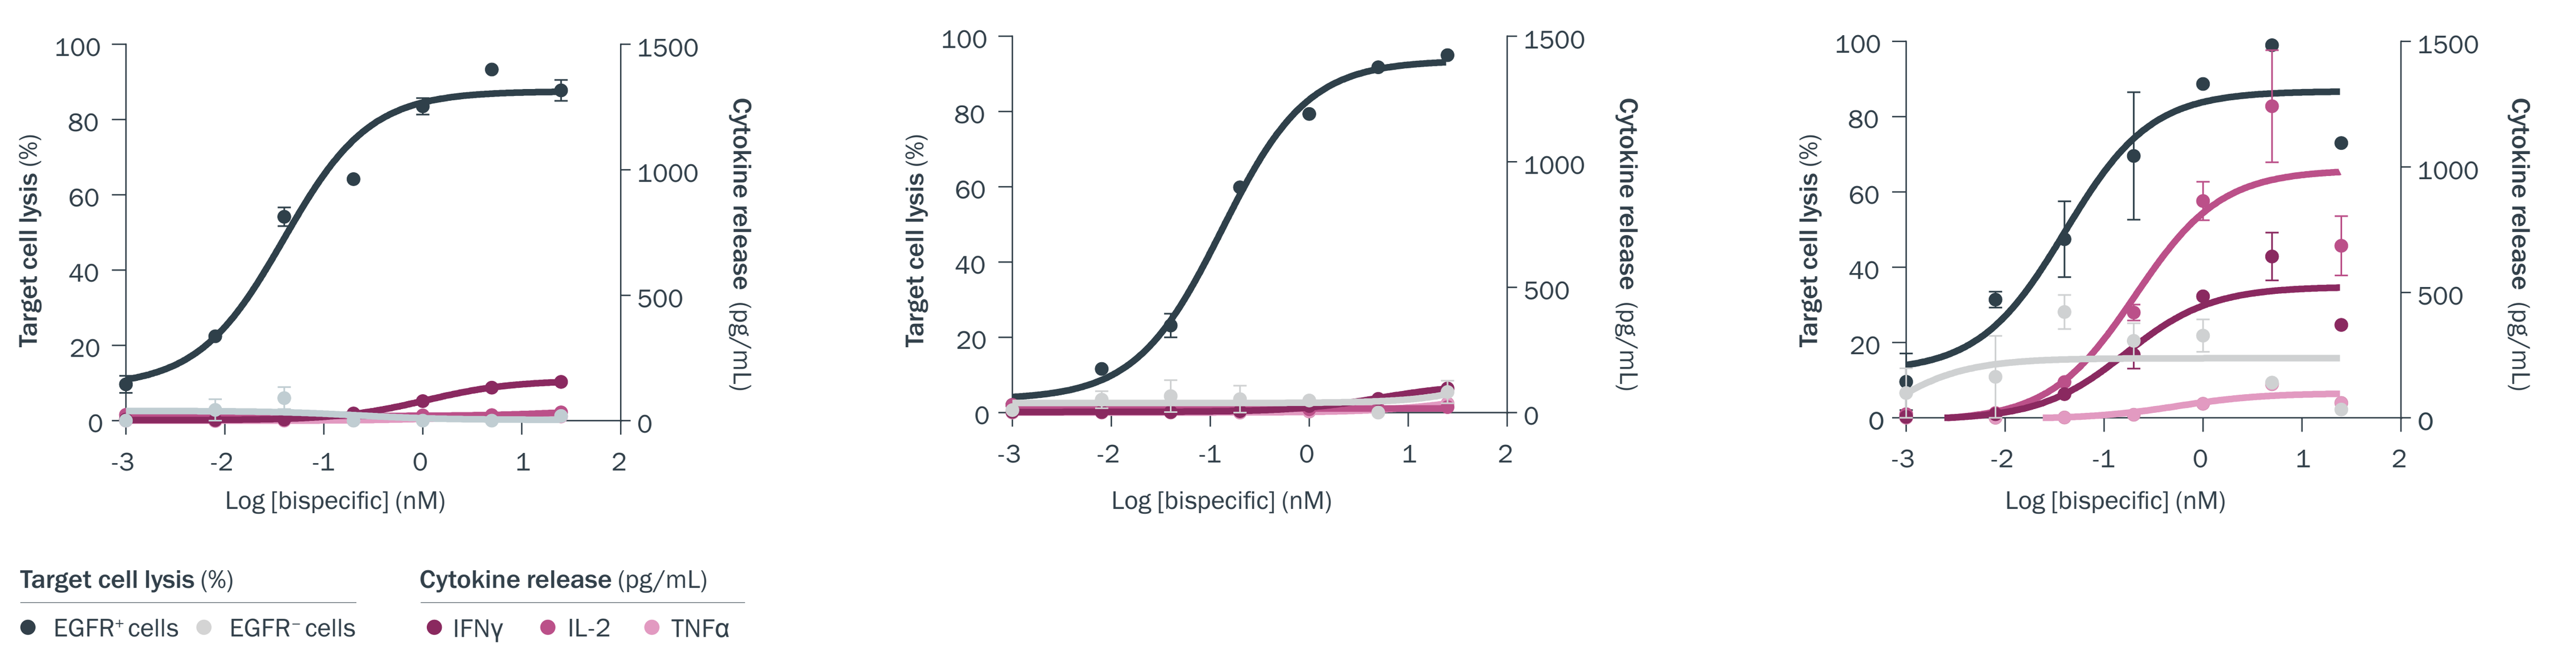
AbCellera CD3-1
AbCellera CD3-2
SP34-derived CD3
Target cell lysis (%)
Cytokine release (pg/mL)
Log [bispecific] (nM)
Target cell lysis (%)
EGFR⁺ cells
EGFR⁻ cells
Cytokine release (pg/mL)
IFNγ
IL-2
TNFα

Figure 2. Functional properties of T-cell engagers engineered with AbCellera's CD3-binding antibodies are superior to benchmark-derived molecules. Tumor cell killing and cytokine release curves were evaluated for CD3 x EGFR antibodies engineered using a fixed EGFR-binding arm (matuzumab) and CD3-binding arms from AbCellera's panel (CD3-1, CD3-2) or pasotuxizumab (SP34-derived) using the assay described in Figure 1.

OrthoMab™-engineered bispecifics with favorable developability properties.

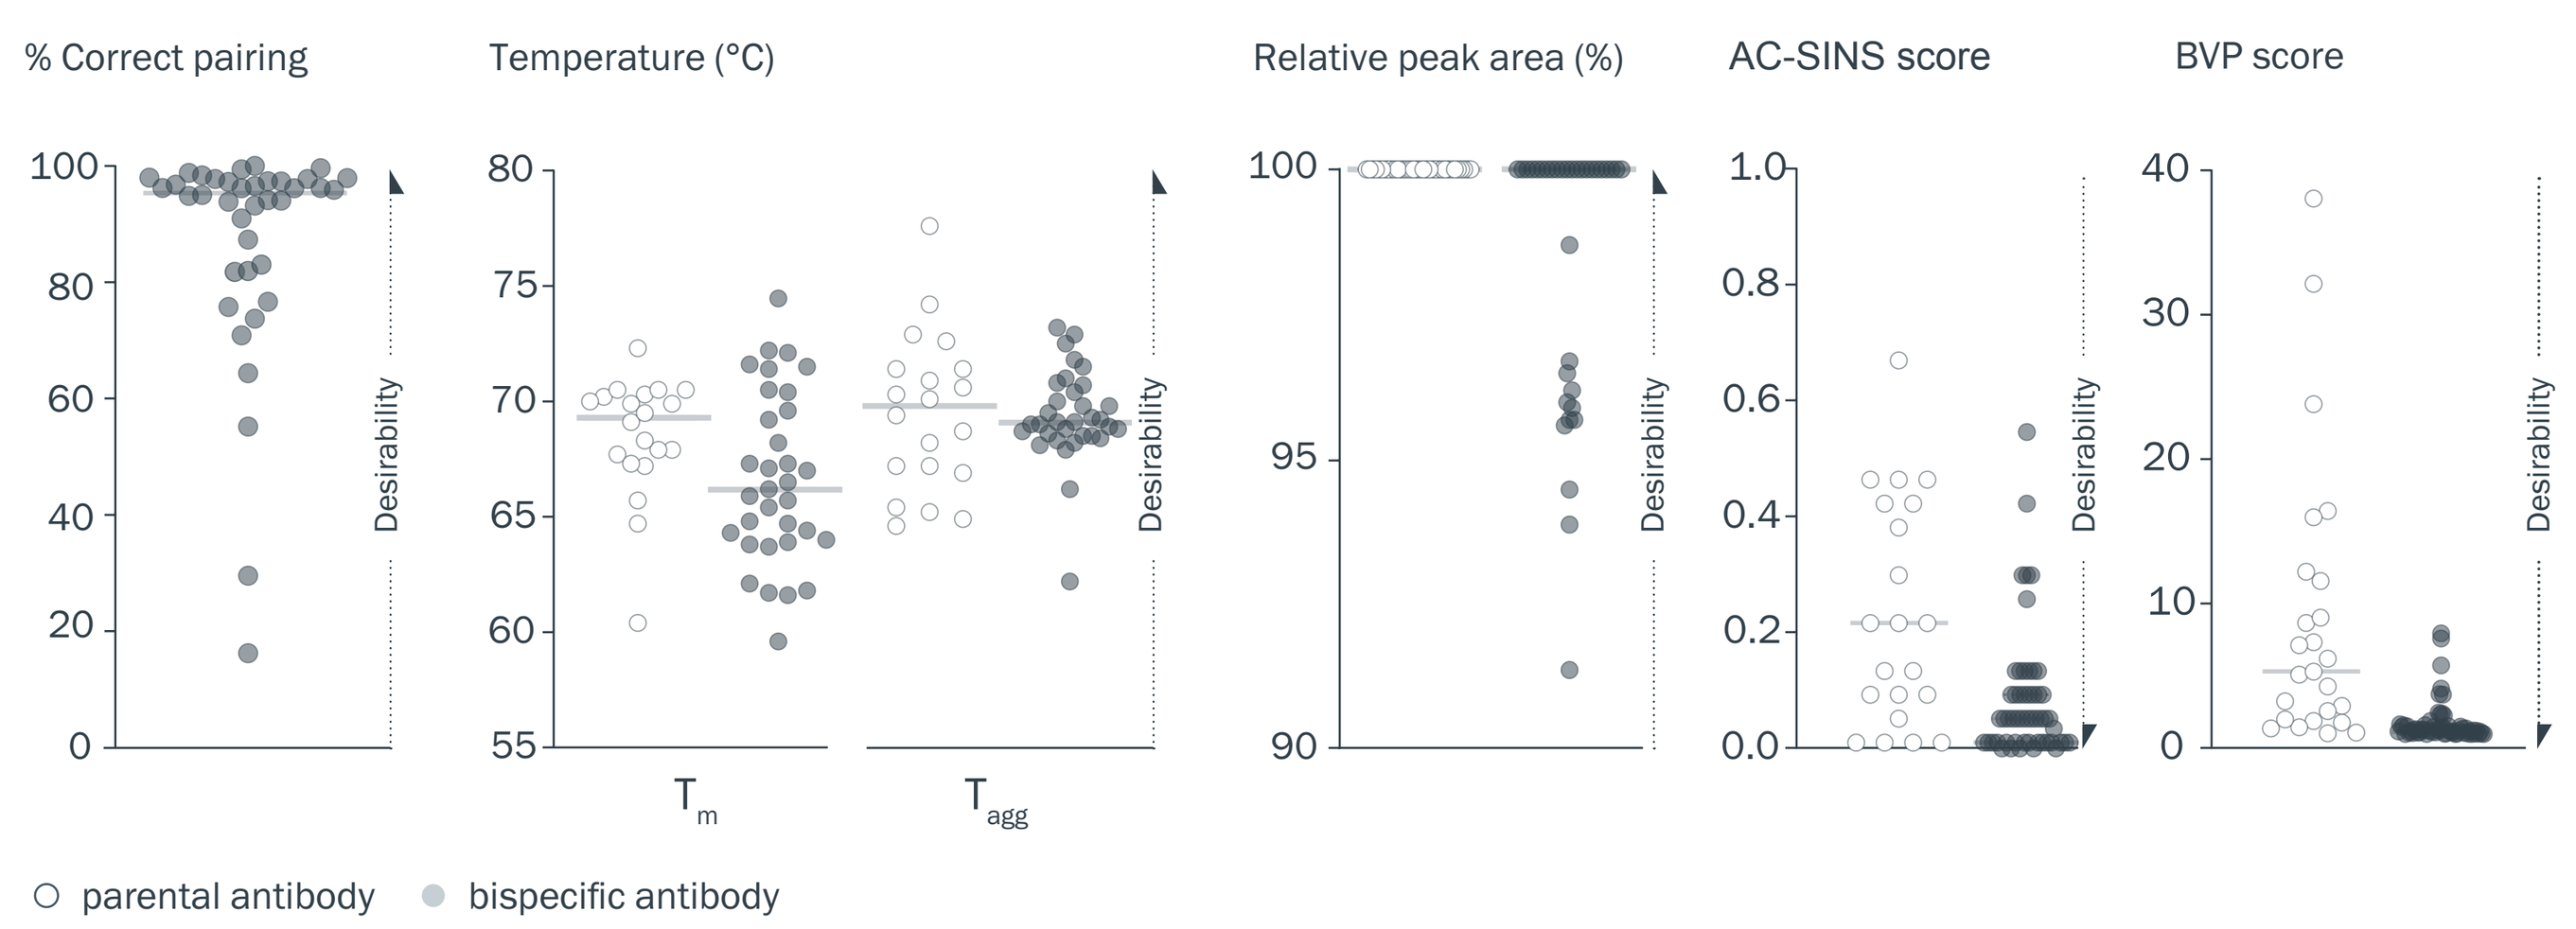
Pairing
Intact MS
% Correct pairing
Stability
nDSF
Temperature (°C)
Tm
Tagg
Monomericity
aSEC
Relative peak area (%)
Self-association
AC-SINS
AC-SINS score
Polyspecificity
BVP-ELISA
BVP score
Desirability
parental antibody
bispecific antibody

Figure 3. Most OrthoMab™-engineered bispecifics have developability properties comparable to parental antibodies. The fractional percentage of correctly paired vs. incorrectly paired bispecifics was measured by intact mass spectrometry (MS). Stability, monomericity, self-association, and polyspecificity were measured by nDSF, aSEC, AC-SINS, and BVP-ELISA, respectively.

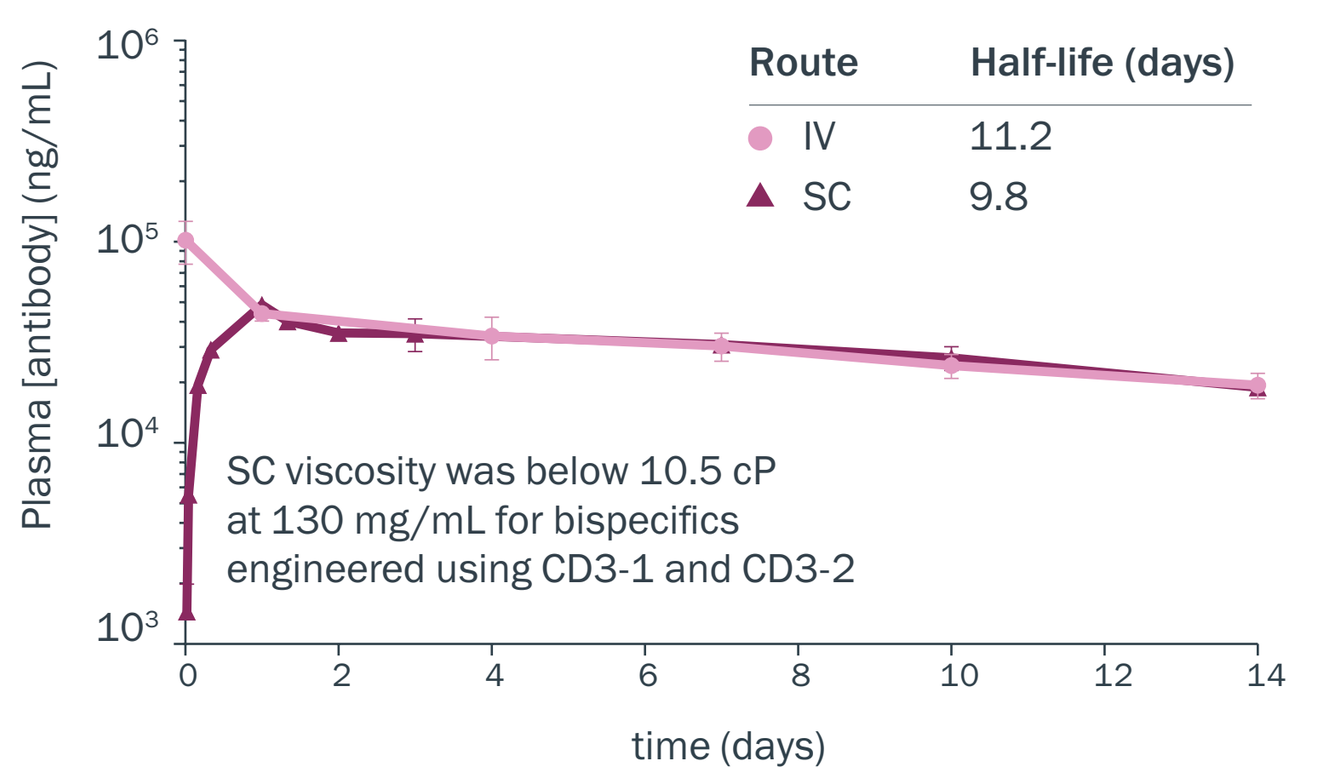
Pharmacokinetics (PK)
Tg32 mice; CD3-1 arm
Plasma [antibody] (ng/mL)
time (days)
Route
Half-life (days)
IV
11.2
SC
9.8
SC viscosity was below 10.5 cP
at 130 mg/mL for bispecifics
engineered using CD3-1 and CD3-2

Figure 4. An exemplary CD3 x EGFR bispecific antibody with favorable PK properties. Following a single 5 mg/kg dose in Tg32 mice (human FcRn transgenic, n=4), concentrations were quantified using an electro-chemiluminescence assay.

Custom-built for different tumor targets

We identified optimal CD3-binding arms for different tumor targets.

T-cell engager function across different tumor targets and target densities

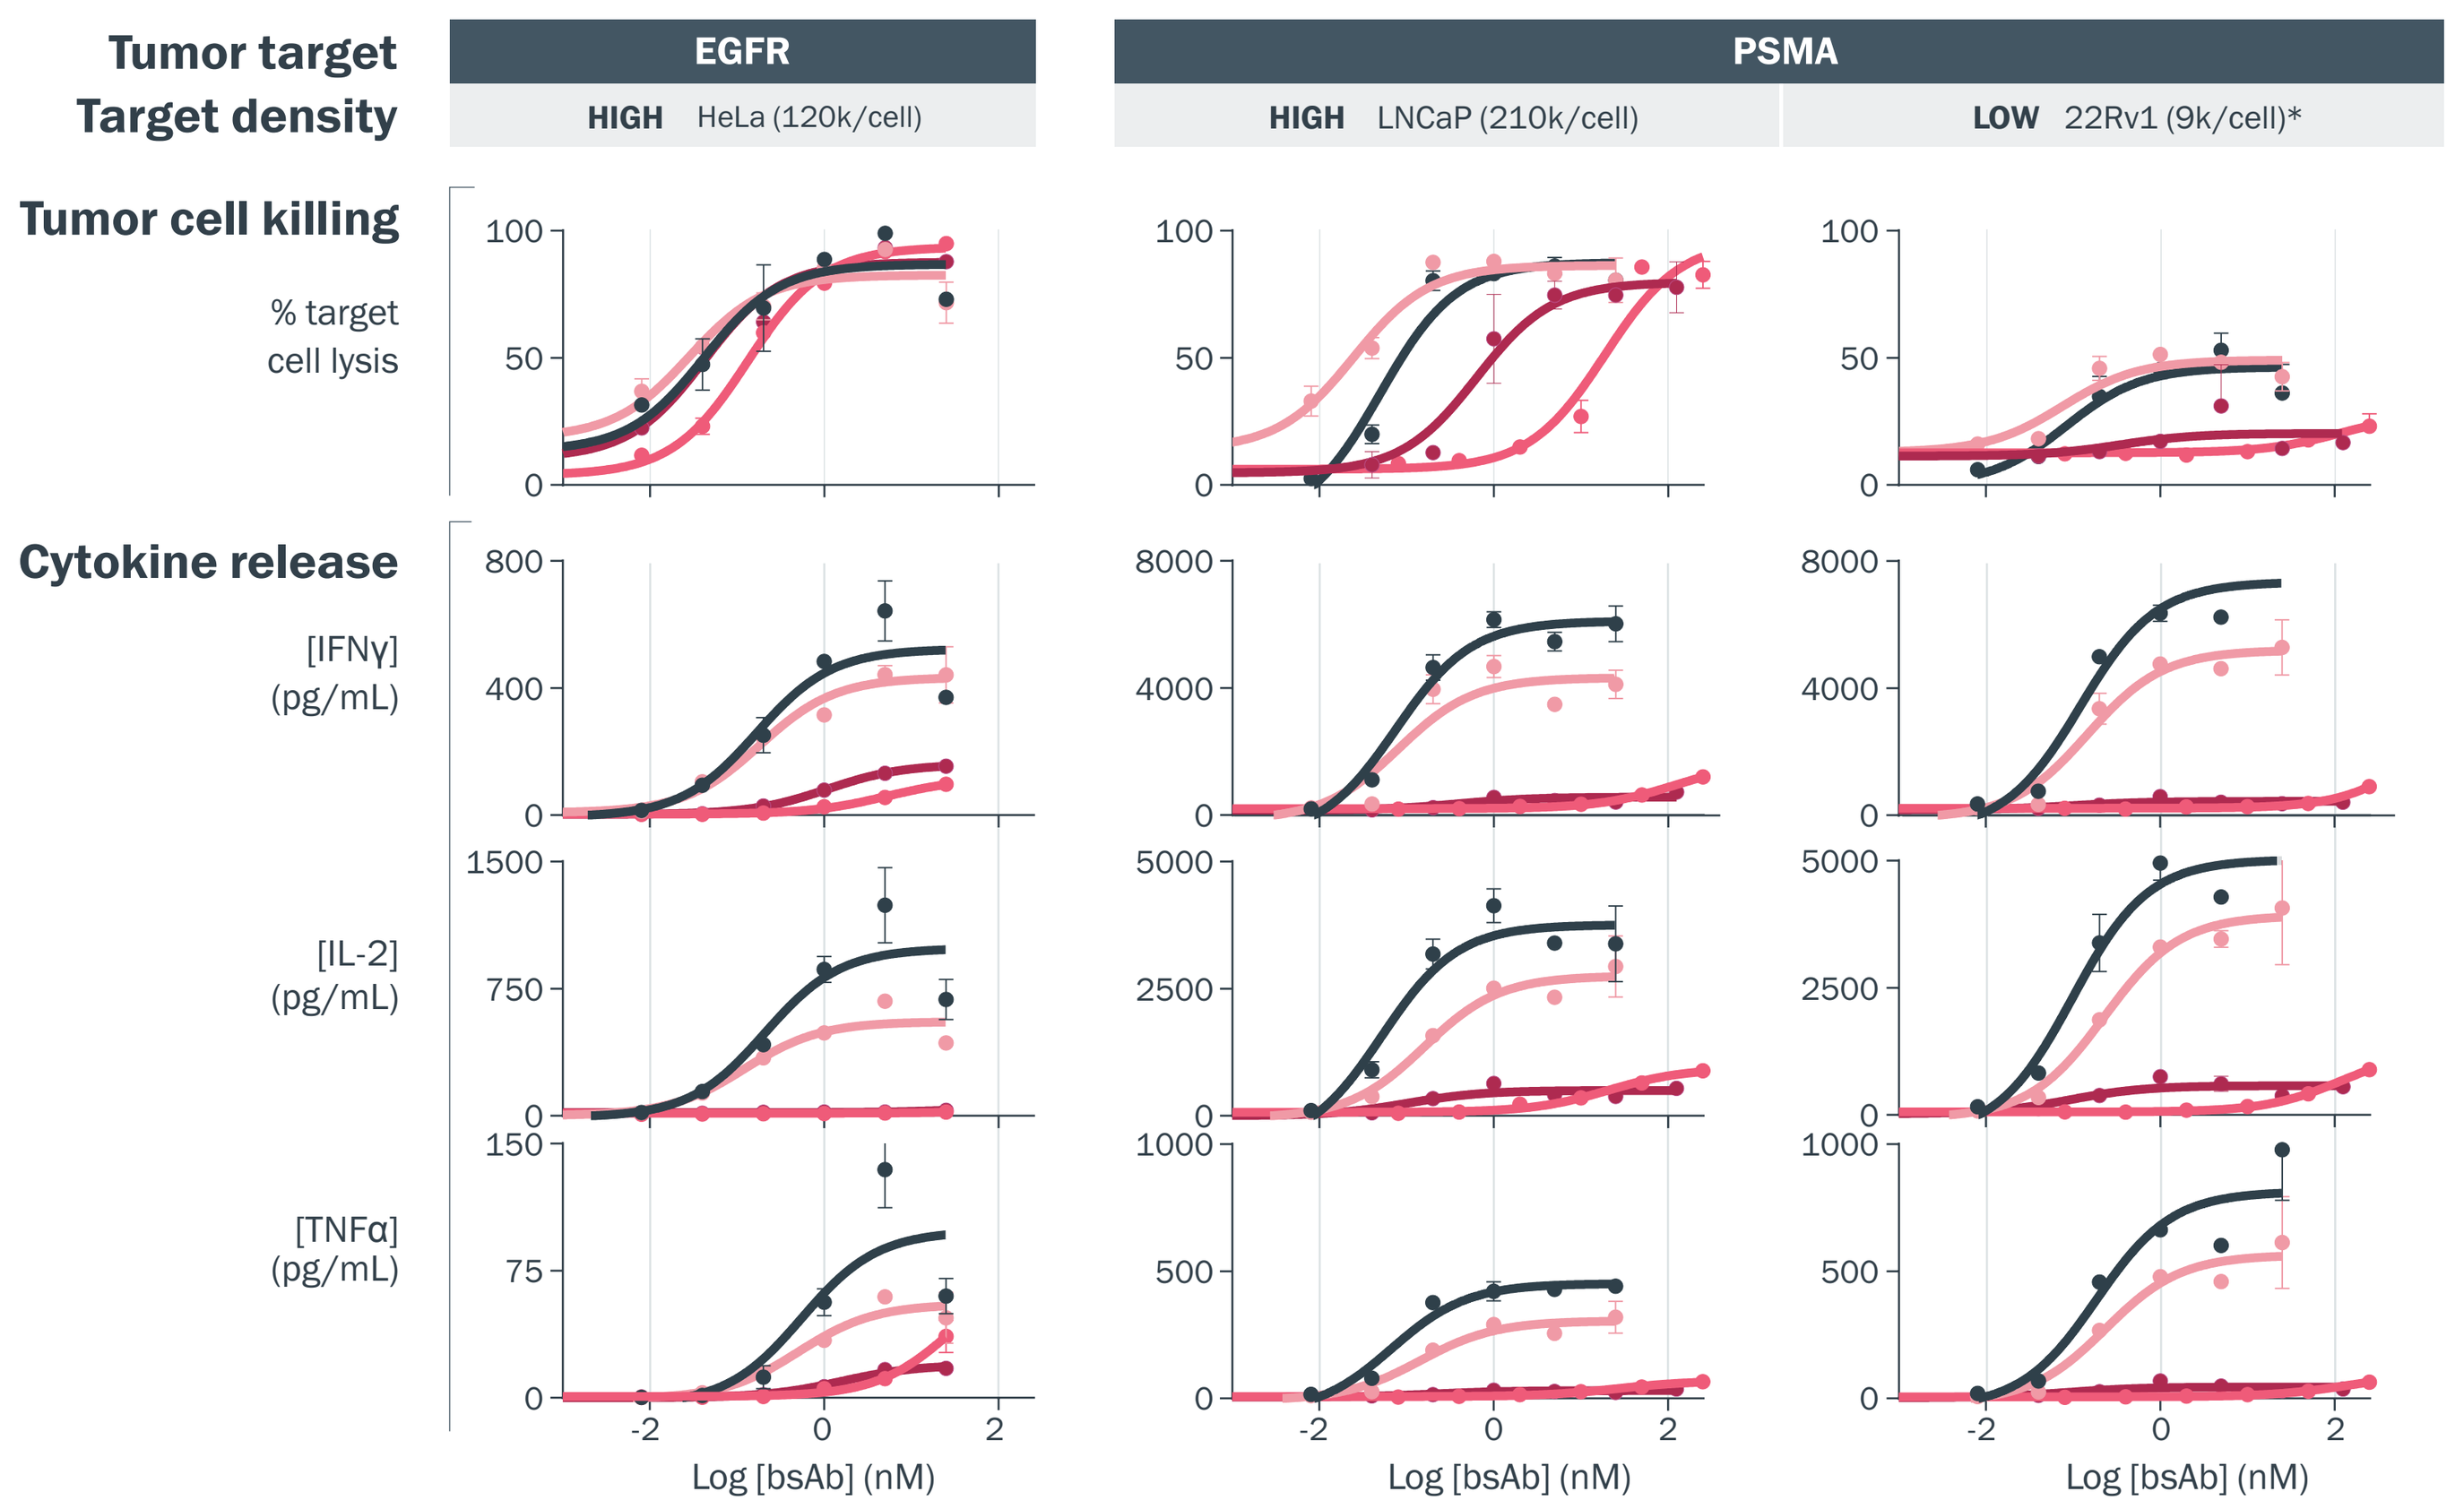
Tumor target
Target density
EGFR
HIGH HeLa (120k/cell)
PSMA
HIGH LNCaP (210k/cell)
LOW 22Rv1 (9k/cell)*
Tumor cell killing
% target cell lysis
Cytokine release
[IFNγ] (pg/mL)
[IL-2] (pg/mL)
[TNFα] (pg/mL)
Log [bsAb] (nM)

Figure 5. The optimal CD3-binding arm is dependent on tumor target and target density. We compared the functional profiles of CD3 x PSMA and CD3 x EGFR T-cell engagers across tumor targets and cell lines. Bispecifics were engineered using CD3-binding arms from AbCellera's panel (CD3-1, CD3-2, CD3-3) or pasotuxizumab, and either a fixed EGFR-binding arm (matuzumab) or a fixed PSMA-binding arm (TNB-585). Functional properties for CD3 x EGFR bispecifics were measured using the assay described in Figure 1. Tumor cell killing and cytokine release for CD3 x PSMA bispecifics were measured 48 hours and 72 hours following addition of the bispecific antibodies for LNCaP and 22Rv1 cells, respectively.

CD3 arms	Affinity (nM)	
AbCellera-discovered		
CD3-1	εδ (351)	εγ(171)
CD3-2	εγ (258)	
CD3-3	εδ (21)	
SP34-derived		
pasotuxizumab		

*Approximately 30% of 22Rv1 cells do not express PSMA (data not shown)

From novel CD3-binding antibodies

Fully human CD3-binding antibodies that are distinct from commonly used molecules.

Antibody sequence diversity overlayed with epitope data

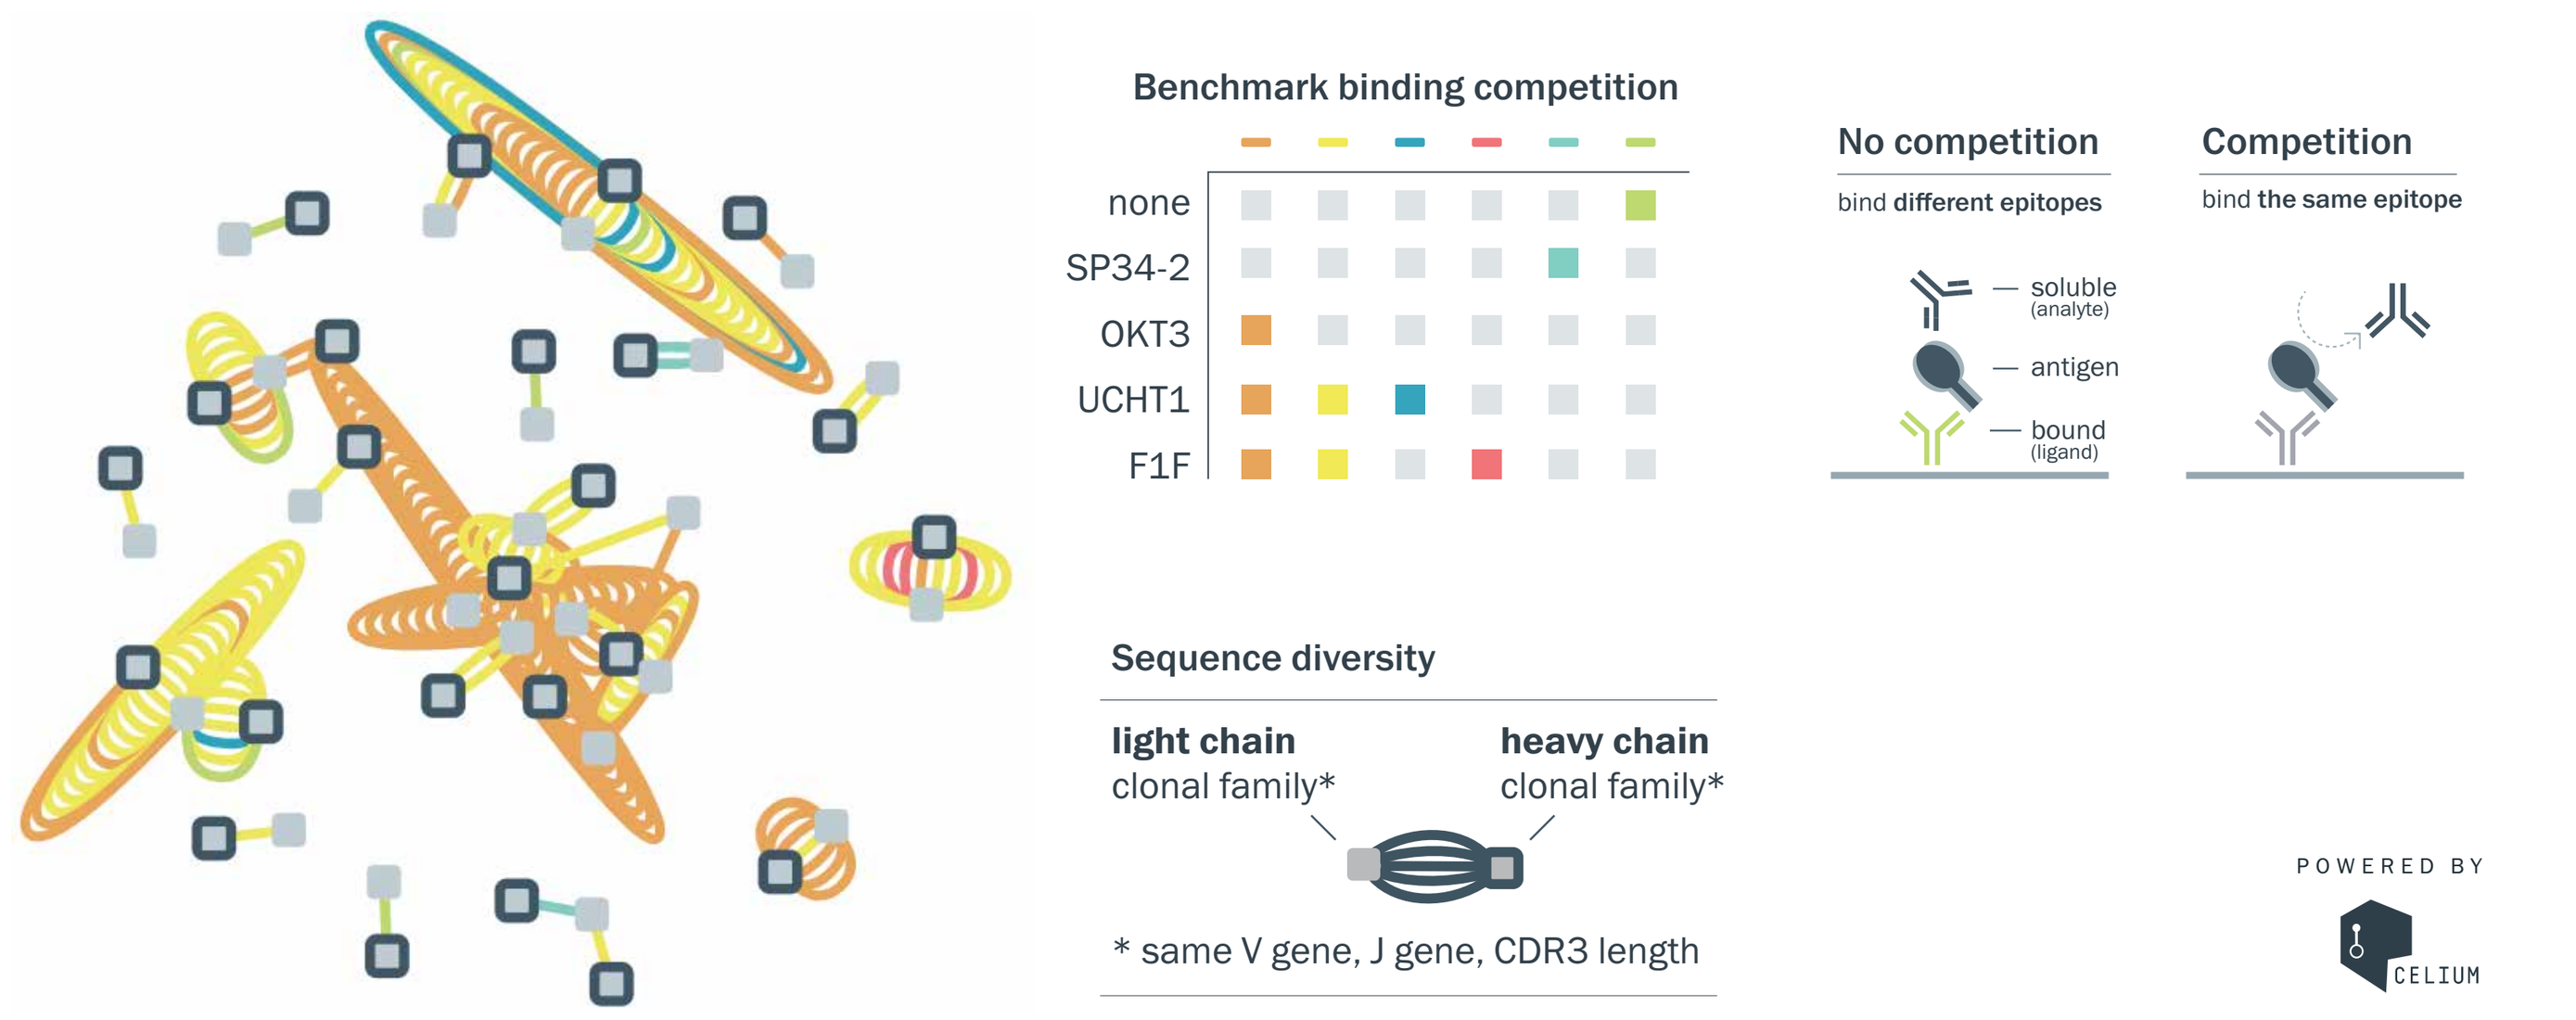
Benchmark binding competition
none
SP34-2
OKT3
UCHT1
F1F
No competition
bind different epitopes
soluble (analyte)
antigen
bound (ligand)
Competition
bind the same epitope
Sequence diversity
light chain
clonal family*
heavy chain
clonal family*
* same V gene, J gene, CDR3 length
POWERED BY
CELIUM

Figure 6. Proprietary immunization protocols using humanized mice yielded CD3-binding antibodies with diverse sequences that bind different epitopes. Antibody sequence diversity was visualized using our proprietary data visualization software, Celium™. Clusters represent clonal families based on V genes, J genes, and CDR3 lengths. The plot is colored using data from high-throughput surface plasmon resonance (SPR) epitope binning, demonstrating that the majority of our CD3-binding antibodies bind epitopes that are distinct from that of SP34-2.

Novel CD3-binding antibodies with diverse binding properties

Broad binding affinities

Human CD3

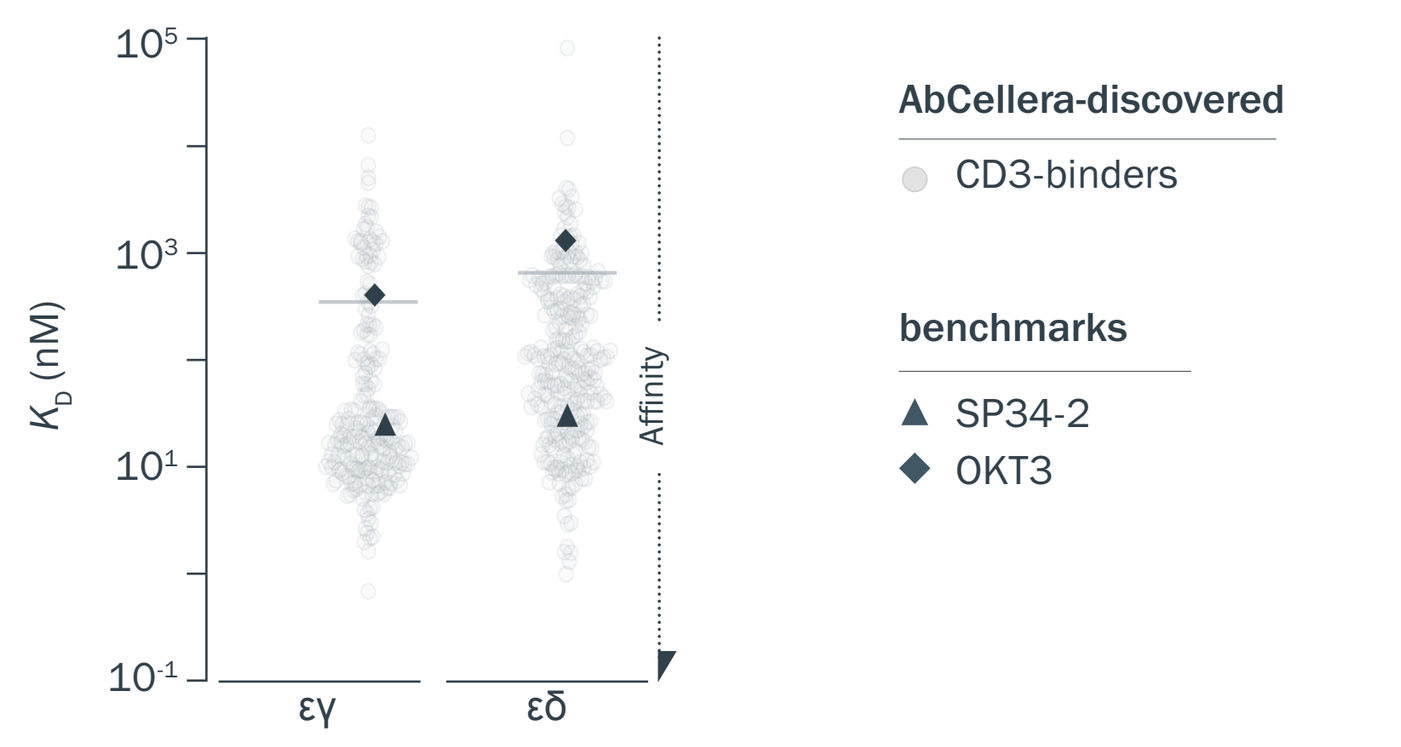
KD (nM)
εγ
εδ
Affinity
AbCellera-discovered
CD3-binders
benchmarks
SP34-2
OKT3

A range of on and off rates

Human CD3εδ

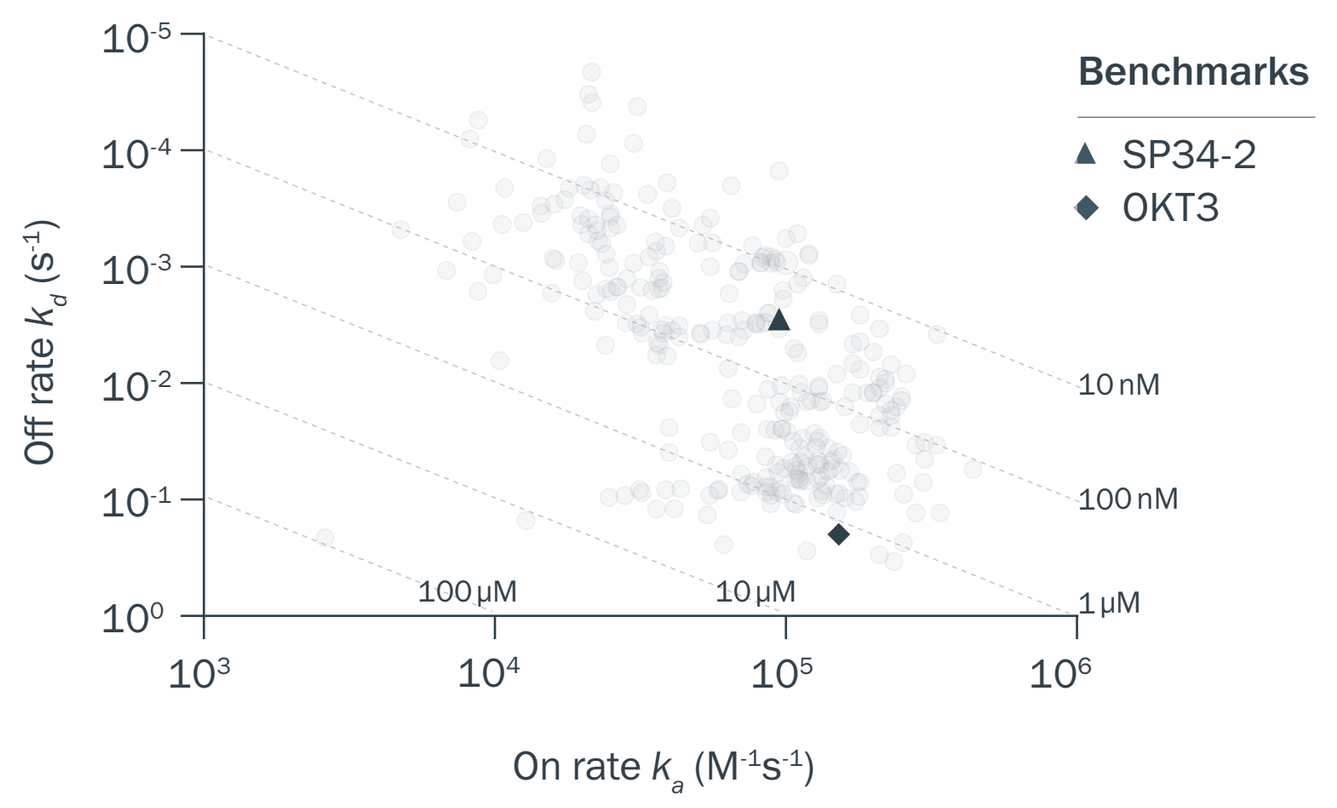
Off rate kd (s⁻¹)
On rate ka (M⁻¹s⁻¹)
Benchmarks
SP34-2
OKT3
10 nM
100 nM
1 μM
10 μM
100 μM

Human + cyno cross-reactive binders

Subunit and species specificities

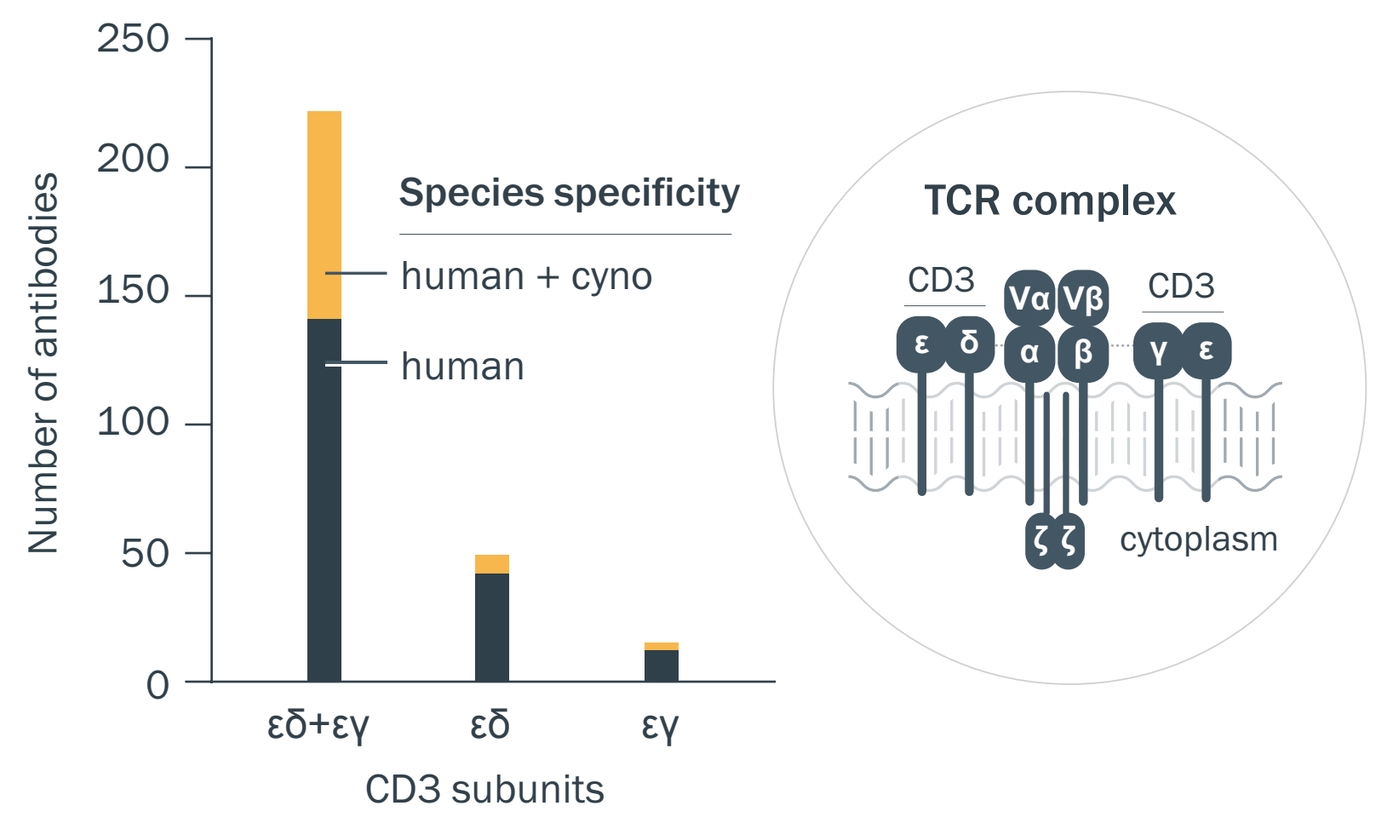
Number of antibodies
CD3 subunits
εδ+εγ
εδ
εγ
Species specificity
human + cyno
human
TCR complex
CD3
cytoplasm

Figure 7. High-throughput assessment of binding properties revealed diverse binding kinetics and specificities. (A) Binding affinity to human CD3 εγ and εδ subunits was measured by SPR-coupled kinetic analysis.

(B) On and off rates were measured by SPR-coupled kinetics and are plotted along the X and Y axes with affinities displayed diagonally.

(C) Subunit specificity was assessed by high-throughput SPR and species specificity was measured by binding to CHO cells expressing either human or cyno CD3.

REFERENCES
1. Pessano, S. et al. 1985. EMBO J. 4(2):337-44.
2. Friedrich, M. et al. 2012. Molecular cancer therapeutics. 11(12):2664-2673.
3. Bacac, M. et al. 2016. Clinical cancer research. 22(13):3286-3297.

COPYRIGHT © ABCELLERA