Functional and specific T-cell engagers for a peptide-MHC tumor target

AUTHORS

Peter Bergqvist, Davide Tortora, Allie Goodman, Claudia Barreto, Ryan Blackler, Kate Caldwell, Lauren Clifford, Stefi Carrara, Lauren Chong, Gabrielle Conaghan, Cindy-Lee Crichlow, Valentine de Puyraimond, Harveer Dhupar, Patrick Farber, Jessica Fernandes Scortecci, Kate Gibson, Rodrigo Goya, Tallie Kuang, Ahn Lee, Franco Li, Tova Pinsky, Craig Robb, Patrick Rowe, Antonios Samiotakis, Eduardo Solano Salgado, **Raffi Tonikian***, Ping Xiang, Irene Yu, Kirstin Brown, Kelly Bullock, Tara Fernandez, Kush Dalal, Stephanie Masterman, Tim Jacobs, Bryan C. Barnhart

**presenter*

AUTHOR AFFILIATION

AbCellera, Vancouver, Canada

DOWNLOAD POSTER

2373

BACKGROUND

pMHCs could expand therapeutic opportunities for T-cell engagers

T-cell engagers (TCEs) are among the most promising new modalities in cancer therapy. However, their target repertoire has been restricted to tumor-associated antigens (TAAs) that are expressed on the cell surface, which make up <15% of cellular proteins.[1,2] Accessing intracellular peptides displayed on MHC class I (pMHCs) would greatly expand the target pool for TCEs.[3]

There are multiple modalities that target pMHCs, including soluble T-cell receptor (TCR)-based molecules and TCR mimic bispecific TCEs. Soluble TCRs must be extensively modified to enhance affinity, which can lead to promiscuous binding independent of the peptide.[4,5] In contrast, antibodies bind pMHCs with affinities in the nanomolar to picomolar range,[6] reducing the engineering required to generate potent molecules. However, structural studies reveal that while TCRs typically bind along the peptide's core,[7] antibodies often bind with a bias towards the termini.[8] This can reduce specificity and necessitates extensive specificity screening to avoid off-target toxicities.

AIM

Generate potent, specific MAGE-A4 x CD3 T-cell engagers

We generated CD3 TCEs targeting melanoma-associated antigen 4 (MAGE-A4)-pMHC, a tumor-specific antigen expressed by many solid tumors, but not by most healthy tissues.[9] We paired six pMHC-binding arms with our diverse CD3-binders[10] and assessed bispecific function. We implemented an *in vitro* and *in silico* workflow to assess specificity of MAGE-A4 x CD3 TCEs across hundreds of pMHCs, and integrated the results with structural data to identify a highly specific molecule that is differentiated from a clinical benchmark.

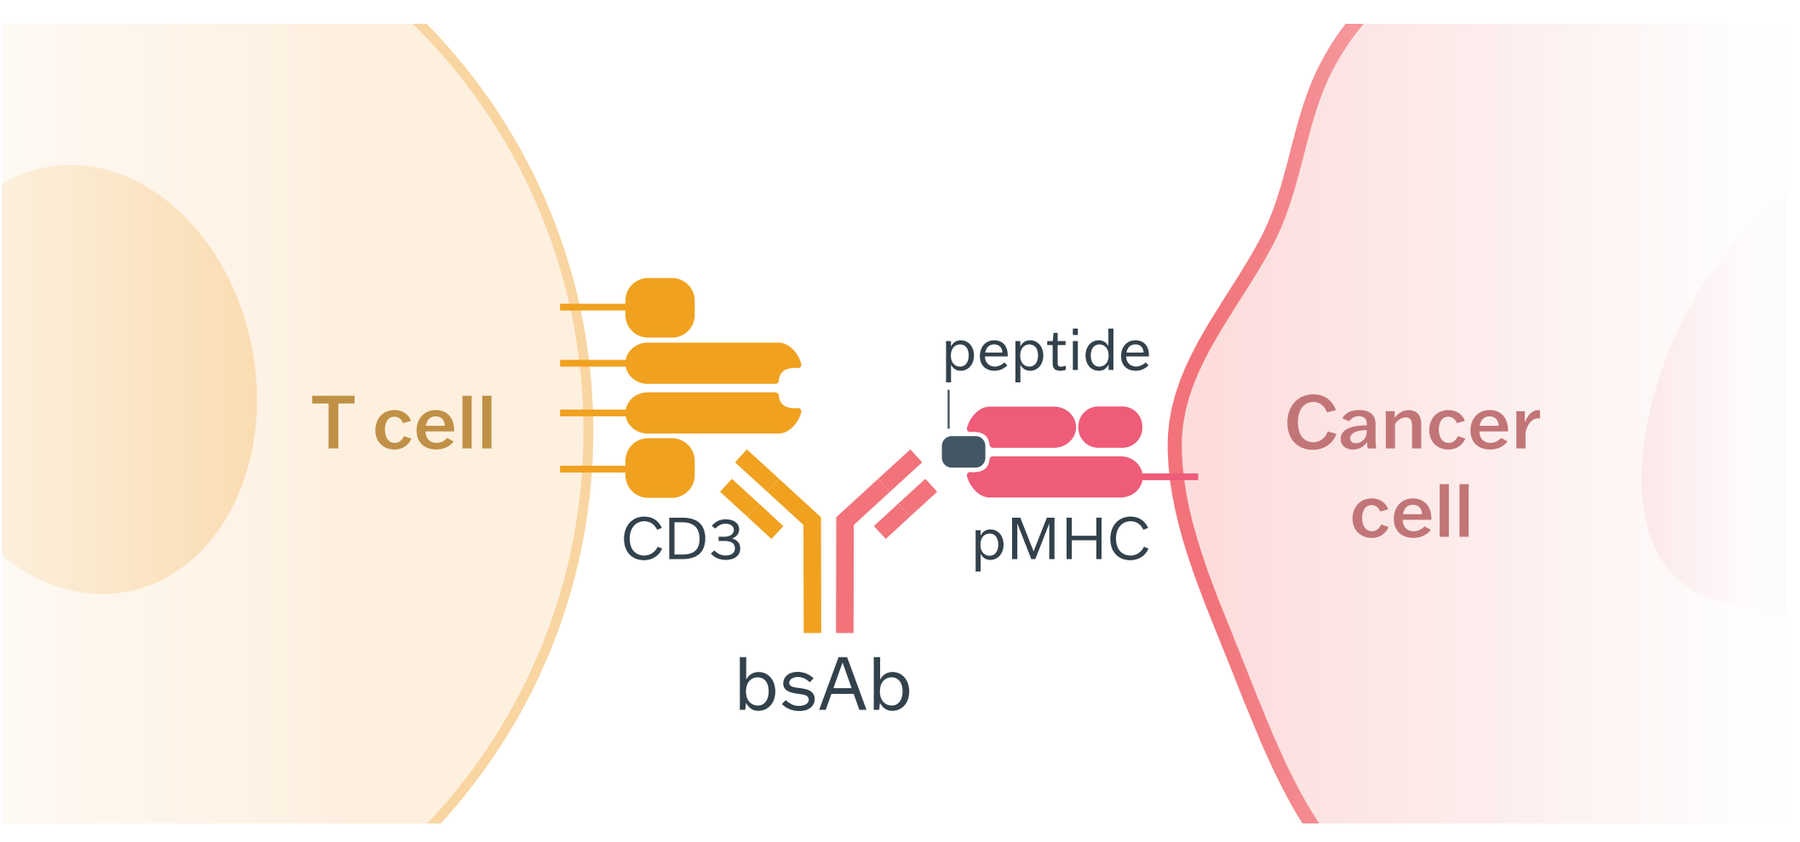

T cell
CD3
peptide
pMHC
bsAb
Cancer cell

Figure 1. Bispecific antibodies (bsAbs) that target pMHCs could expand therapeutic opportunities for TCEs.

OUTCOME

A MAGE-A4 x CD3 T-cell engager that is differentiated from a clinical benchmark

We identified a potent, highly specific MAGE-A4 x CD3 TCE for further preclinical assessment that shows:

- potent activity across multiple MAGE-A4-expressing cell lines with no activity against MAGE-A4-negative cells
- highly specific binding to MAGE-A4-pMHC with no binding to more than 180 non-MAGE-A4 pMHCs
- binding predominantly to the central residues of the MAGE-A4 peptide

MAGE-A4 x CD3 T-cell engagers with potent and specific tumor-cell killing

Bispecific 1 and 2 show target-specific tumor-cell killing across multiple cell lines

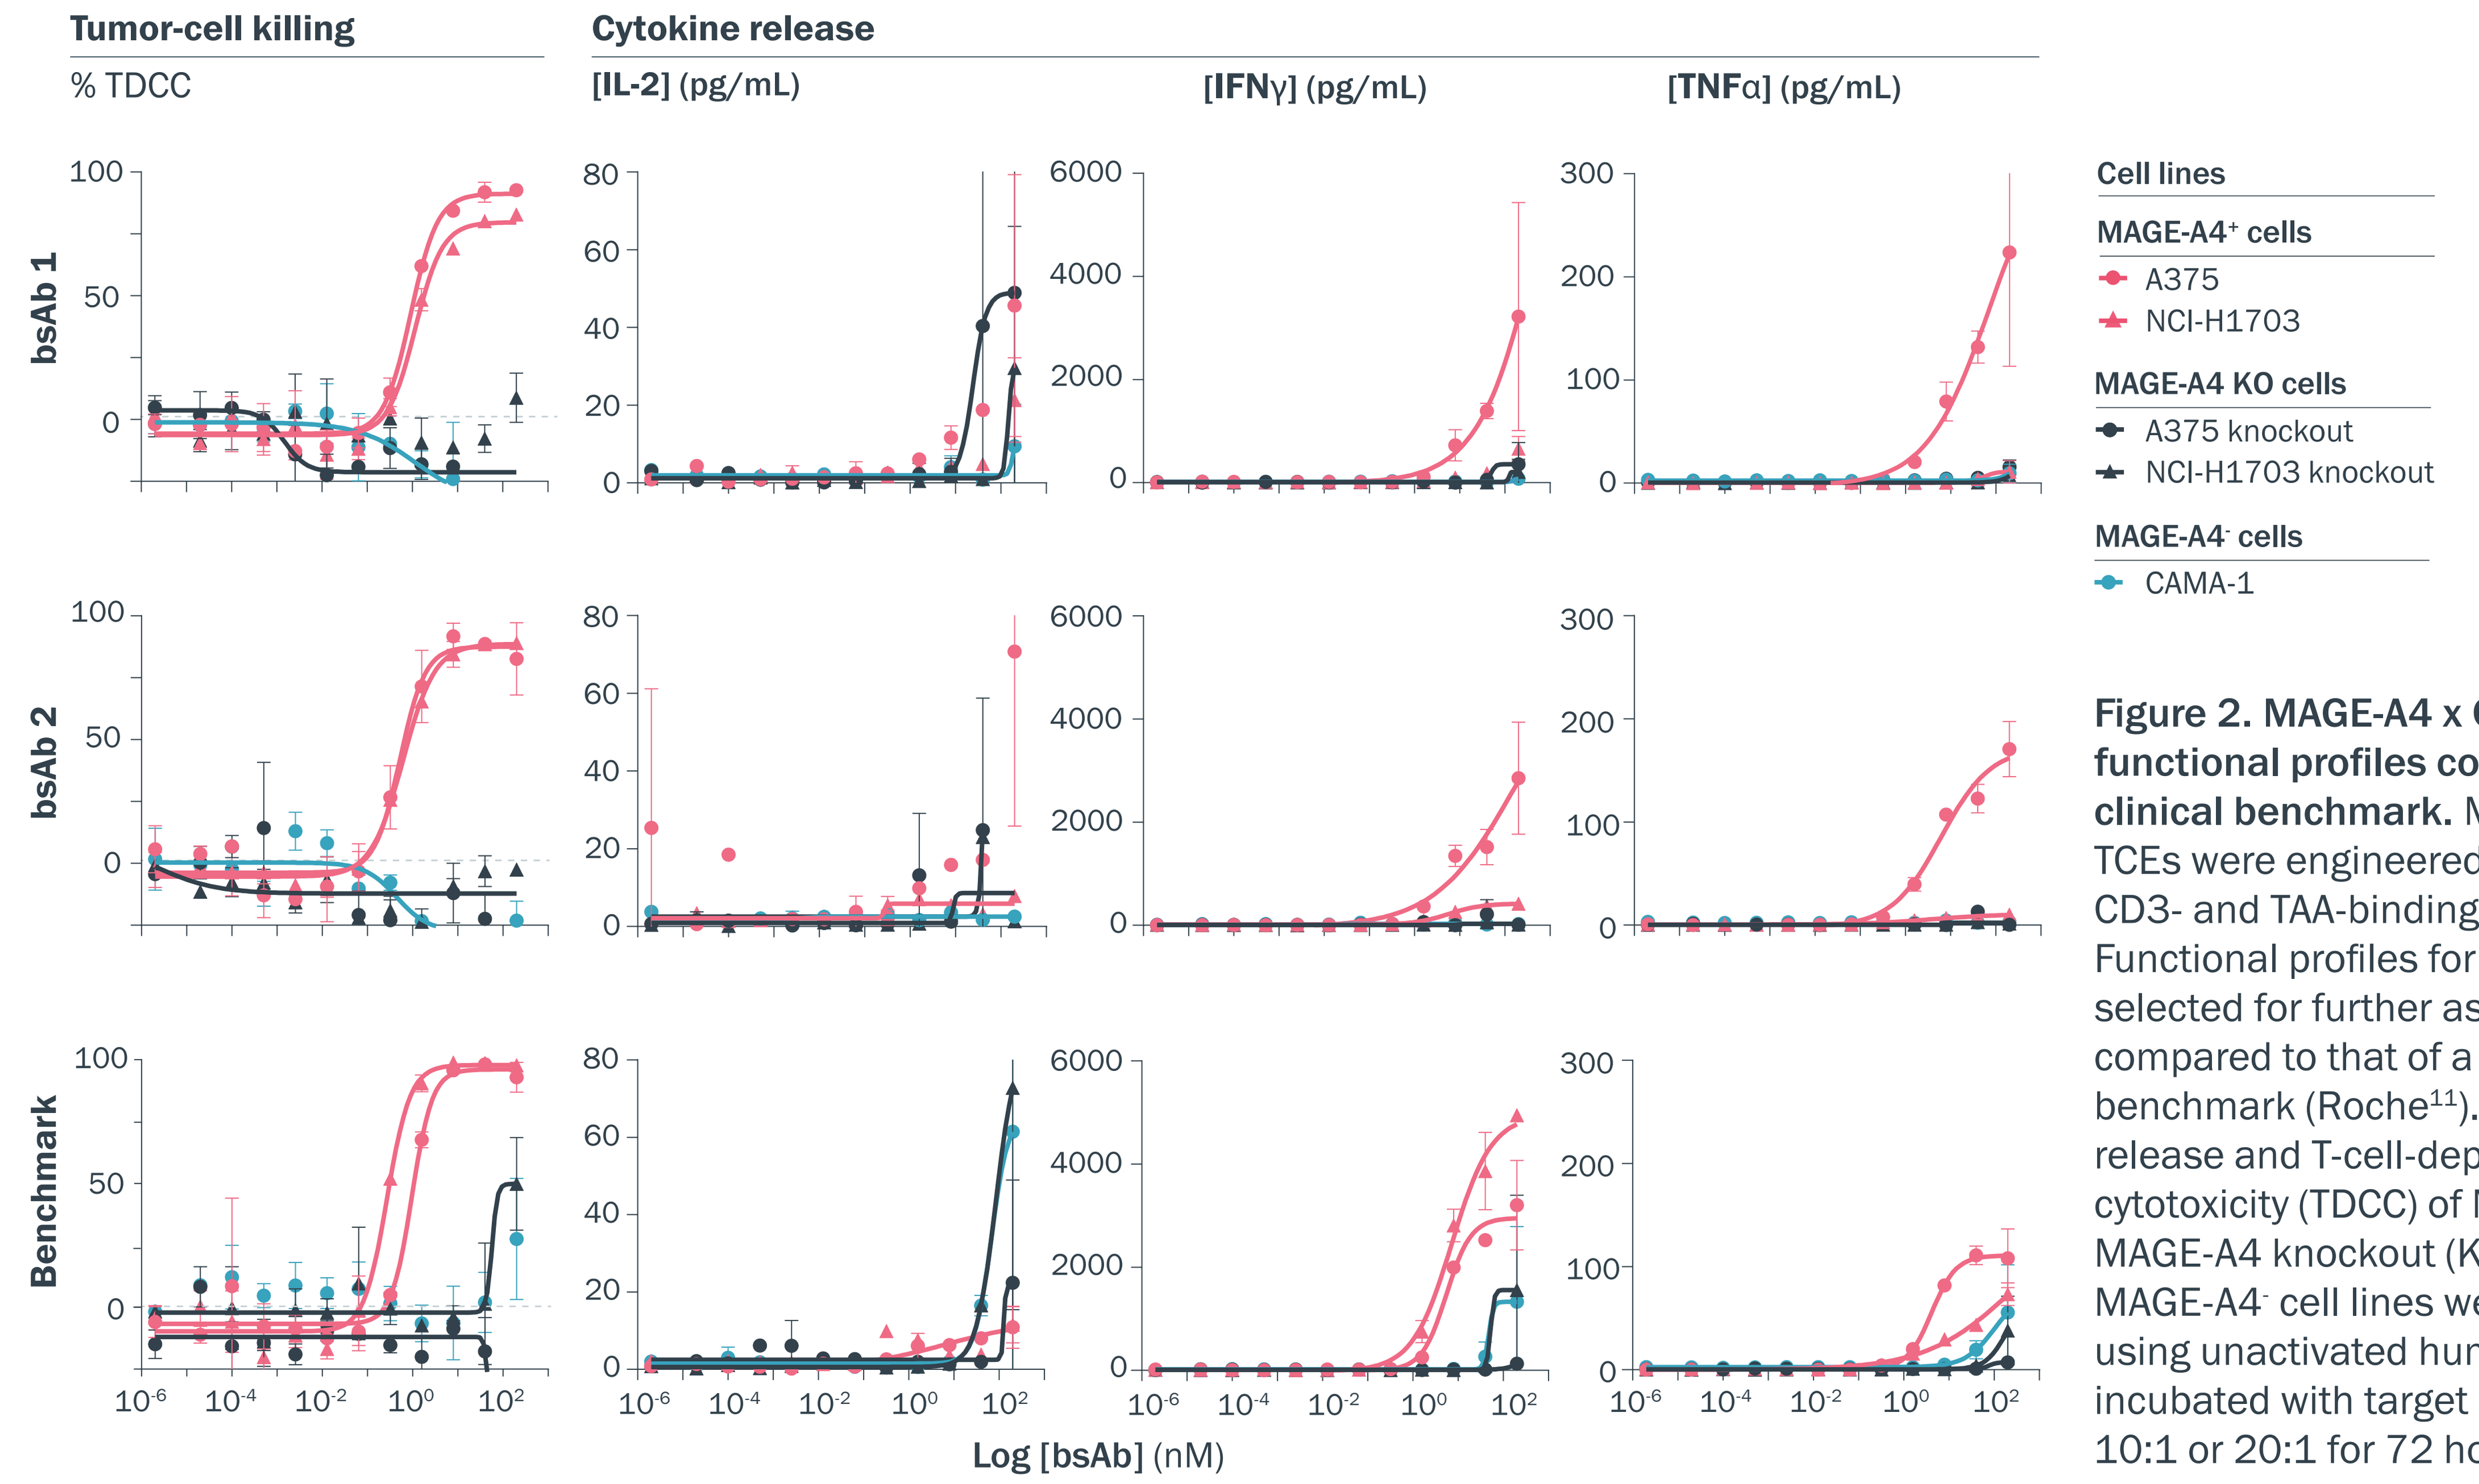

Tumor-cell killing
% TDCC
Cytokine release
[IL-2] (pg/mL)
[IFNγ] (pg/mL)
[TNFα] (pg/mL)
bsAb 1
bsAb 2
Benchmark
Log [bsAb] (nM)
Cell lines
MAGE-A4+ cells
A375
NCI-H1703
MAGE-A4 KO cells
A375 knockout
NCI-H1703 knockout
MAGE-A4- cells
CAMA-1

Figure 2. MAGE-A4 x CD3 TCEs show functional profiles comparable to a clinical benchmark. More than 200 TCEs were engineered using diverse CD3- and TAA-binding arms. Functional profiles for two molecules selected for further assessment are compared to that of a clinical benchmark (Roche[11]). Cytokine release and T-cell-dependent cellular cytotoxicity (TDCC) of MAGE-A4$^+$, MAGE-A4 knockout (KO), and MAGE-A4$^-$ cell lines were measured using unactivated human T cells incubated with target cells at a ratio of 10:1 or 20:1 for 72 hours.

A T-cell engager with high specificity for MAGE-A4-pMHC

Bispecific 1 does not show binding to any of the 180$^+$ non-MAGE peptides tested

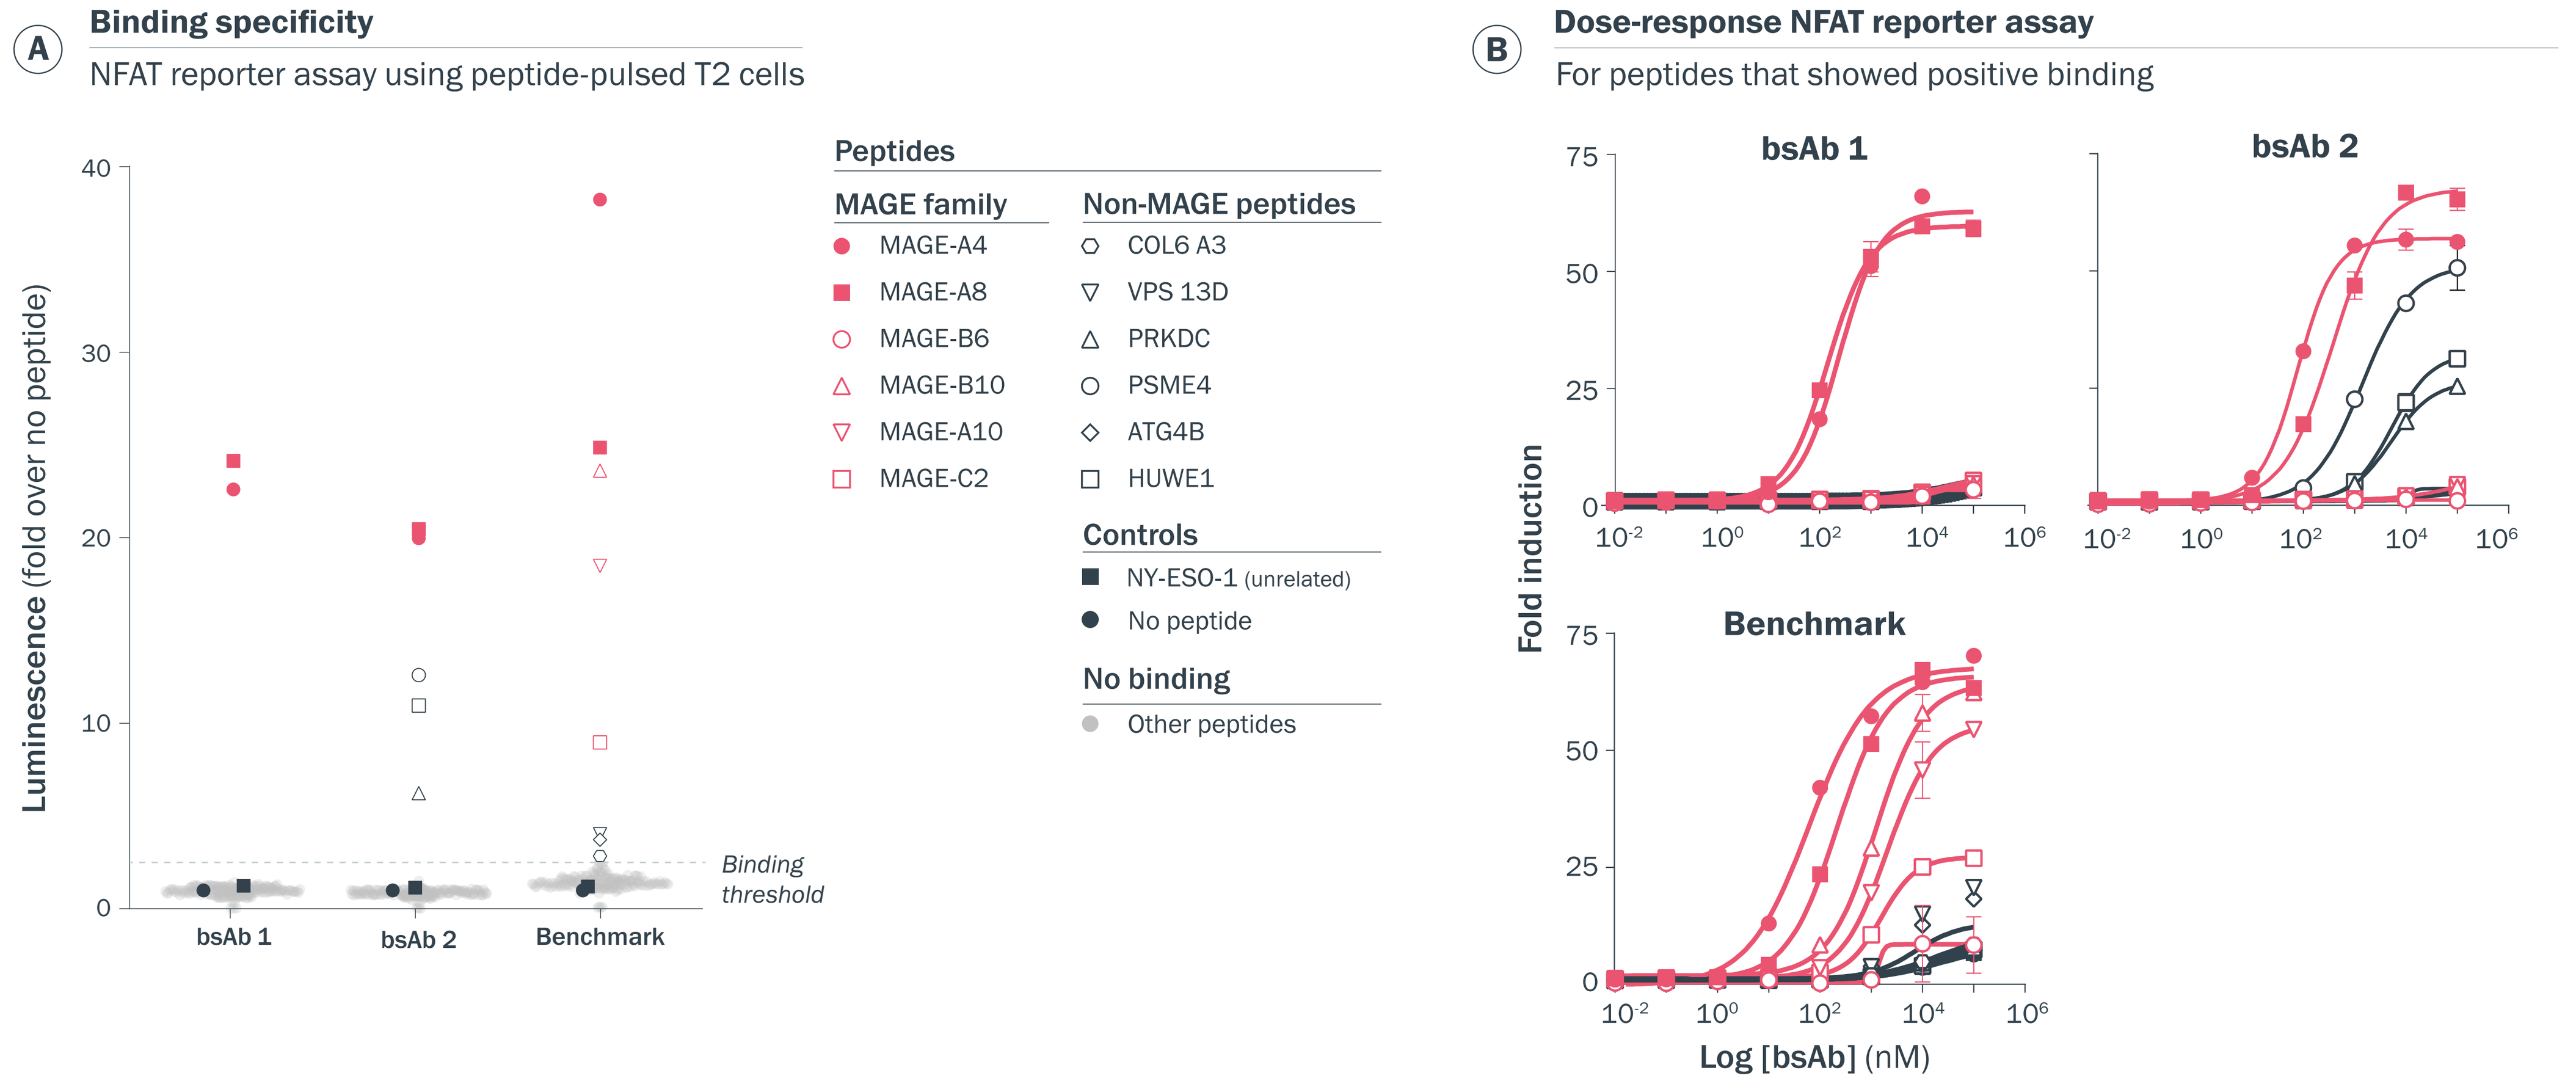

A
Binding specificity
NFAT reporter assay using peptide-pulsed T2 cells
Luminescence (fold over no peptide)
bsAb 1
bsAb 2
Benchmark
Binding threshold
Peptides
MAGE family
MAGE-A4
MAGE-A8
MAGE-B6
MAGE-B10
MAGE-A10
MAGE-C2
Non-MAGE peptides
COL6 A3
VPS 13D
PRKDC
PSME4
ATG4B
HUWE1
Controls
NY-ESO-1 (unrelated)
No peptide
No binding
Other peptides
B
Dose-response NFAT reporter assay
For peptides that showed positive binding
Fold induction
bsAb 1
bsAb 2
Benchmark
Log [bsAb] (nM)

Figure 3. Specificity assessment shows that Bispecific 1 specifically binds tumor-associated peptides from MAGE-A4- and MAGE-A8-pMHC but not to any other peptides tested. Antibody-pMHC binding data from X-scan and structural analyses (Fig. 4) were integrated into the CrossDome package[12] to select peptides for antibody specificity assessment. **(A)** T2 cells were pulsed with relevant peptides and co-cultured with bispecifics and Jurkat NFAT reporter cells. Activation of the NFAT response element indicates positive binding to the peptide-pulsed T2 cells. **(B)** Dose-response curves were generated for peptides showing a signal greater than 2.5-fold over the no peptide control to validate single-point data shown in Fig. 3A.

pMHC-binding profiles that are differentiated from a clinical benchmark

Antibody-pMHC interactions profiled using structural and substitution analyses

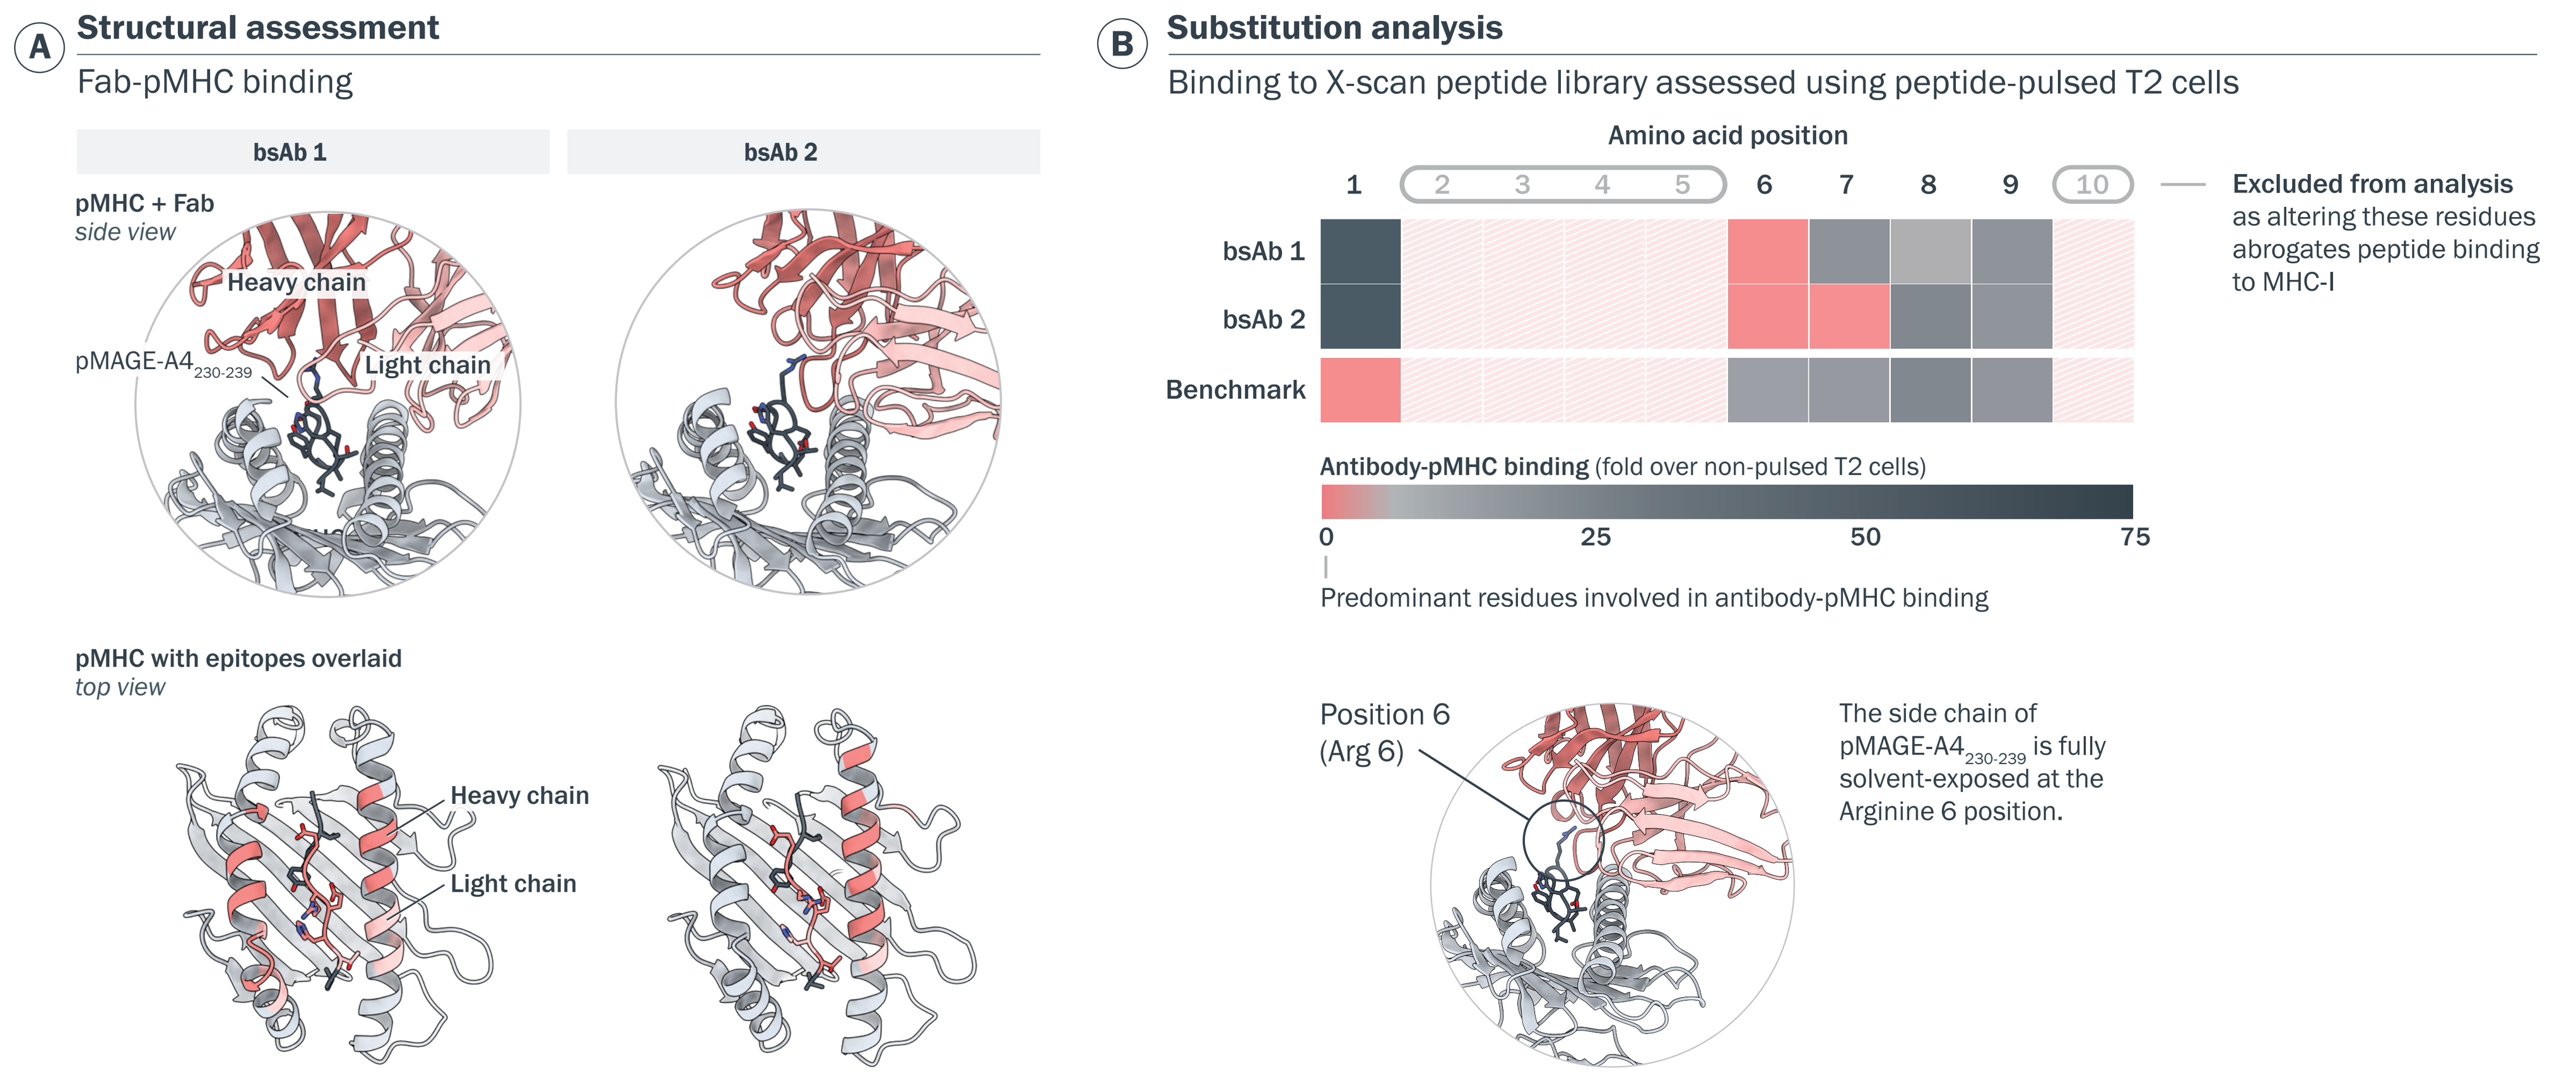

A
Structural assessment
Fab-pMHC binding
bsAb 1
bsAb 2
pMHC + Fab
side view
Heavy chain
Light chain
pMAGE-A4 230-239
pMHC with epitopes overlaid
top view
B
Substitution analysis
Binding to X-scan peptide library assessed using peptide-pulsed T2 cells
Amino acid position
Excluded from analysis as altering these residues abrogates peptide binding to MHC-I
bsAb 1
bsAb 2
Benchmark
Antibody-pMHC binding (fold over non-pulsed T2 cells)
Predominant residues involved in antibody-pMHC binding
Position 6 (Arg 6)
The side chain of pMAGE-A4 230-239 is fully solvent-exposed at the Arginine 6 position.

Figure 4. pMHC binding properties show differentiation from a clinical benchmark. (A) Antibody Fabs bound to pMAGE-A4$_{230\text{-}239}$ displayed on MHC-I (HLA-A*02:01) were assessed by cryo-electron microscopy at 2.7 to 3.3 Å and show peptide-centric binding. **(B)** Each amino acid of pMAGE-A4$_{230\text{-}239}$ was replaced with every possible amino acid to generate 190 variants. Substitutions that abrogated peptide binding to MHC-I (assessed by flow cytometry) were excluded from the analysis. The median values of antibody-pMHC binding compared to the benchmark (Molecule R, monospecific IgG format)[11] are shown.

Generation of rare potent and specific T-cell engagers for pMHC targets

A high-throughput platform generated pMHC-targeting T-cell engagers without extensive protein engineering

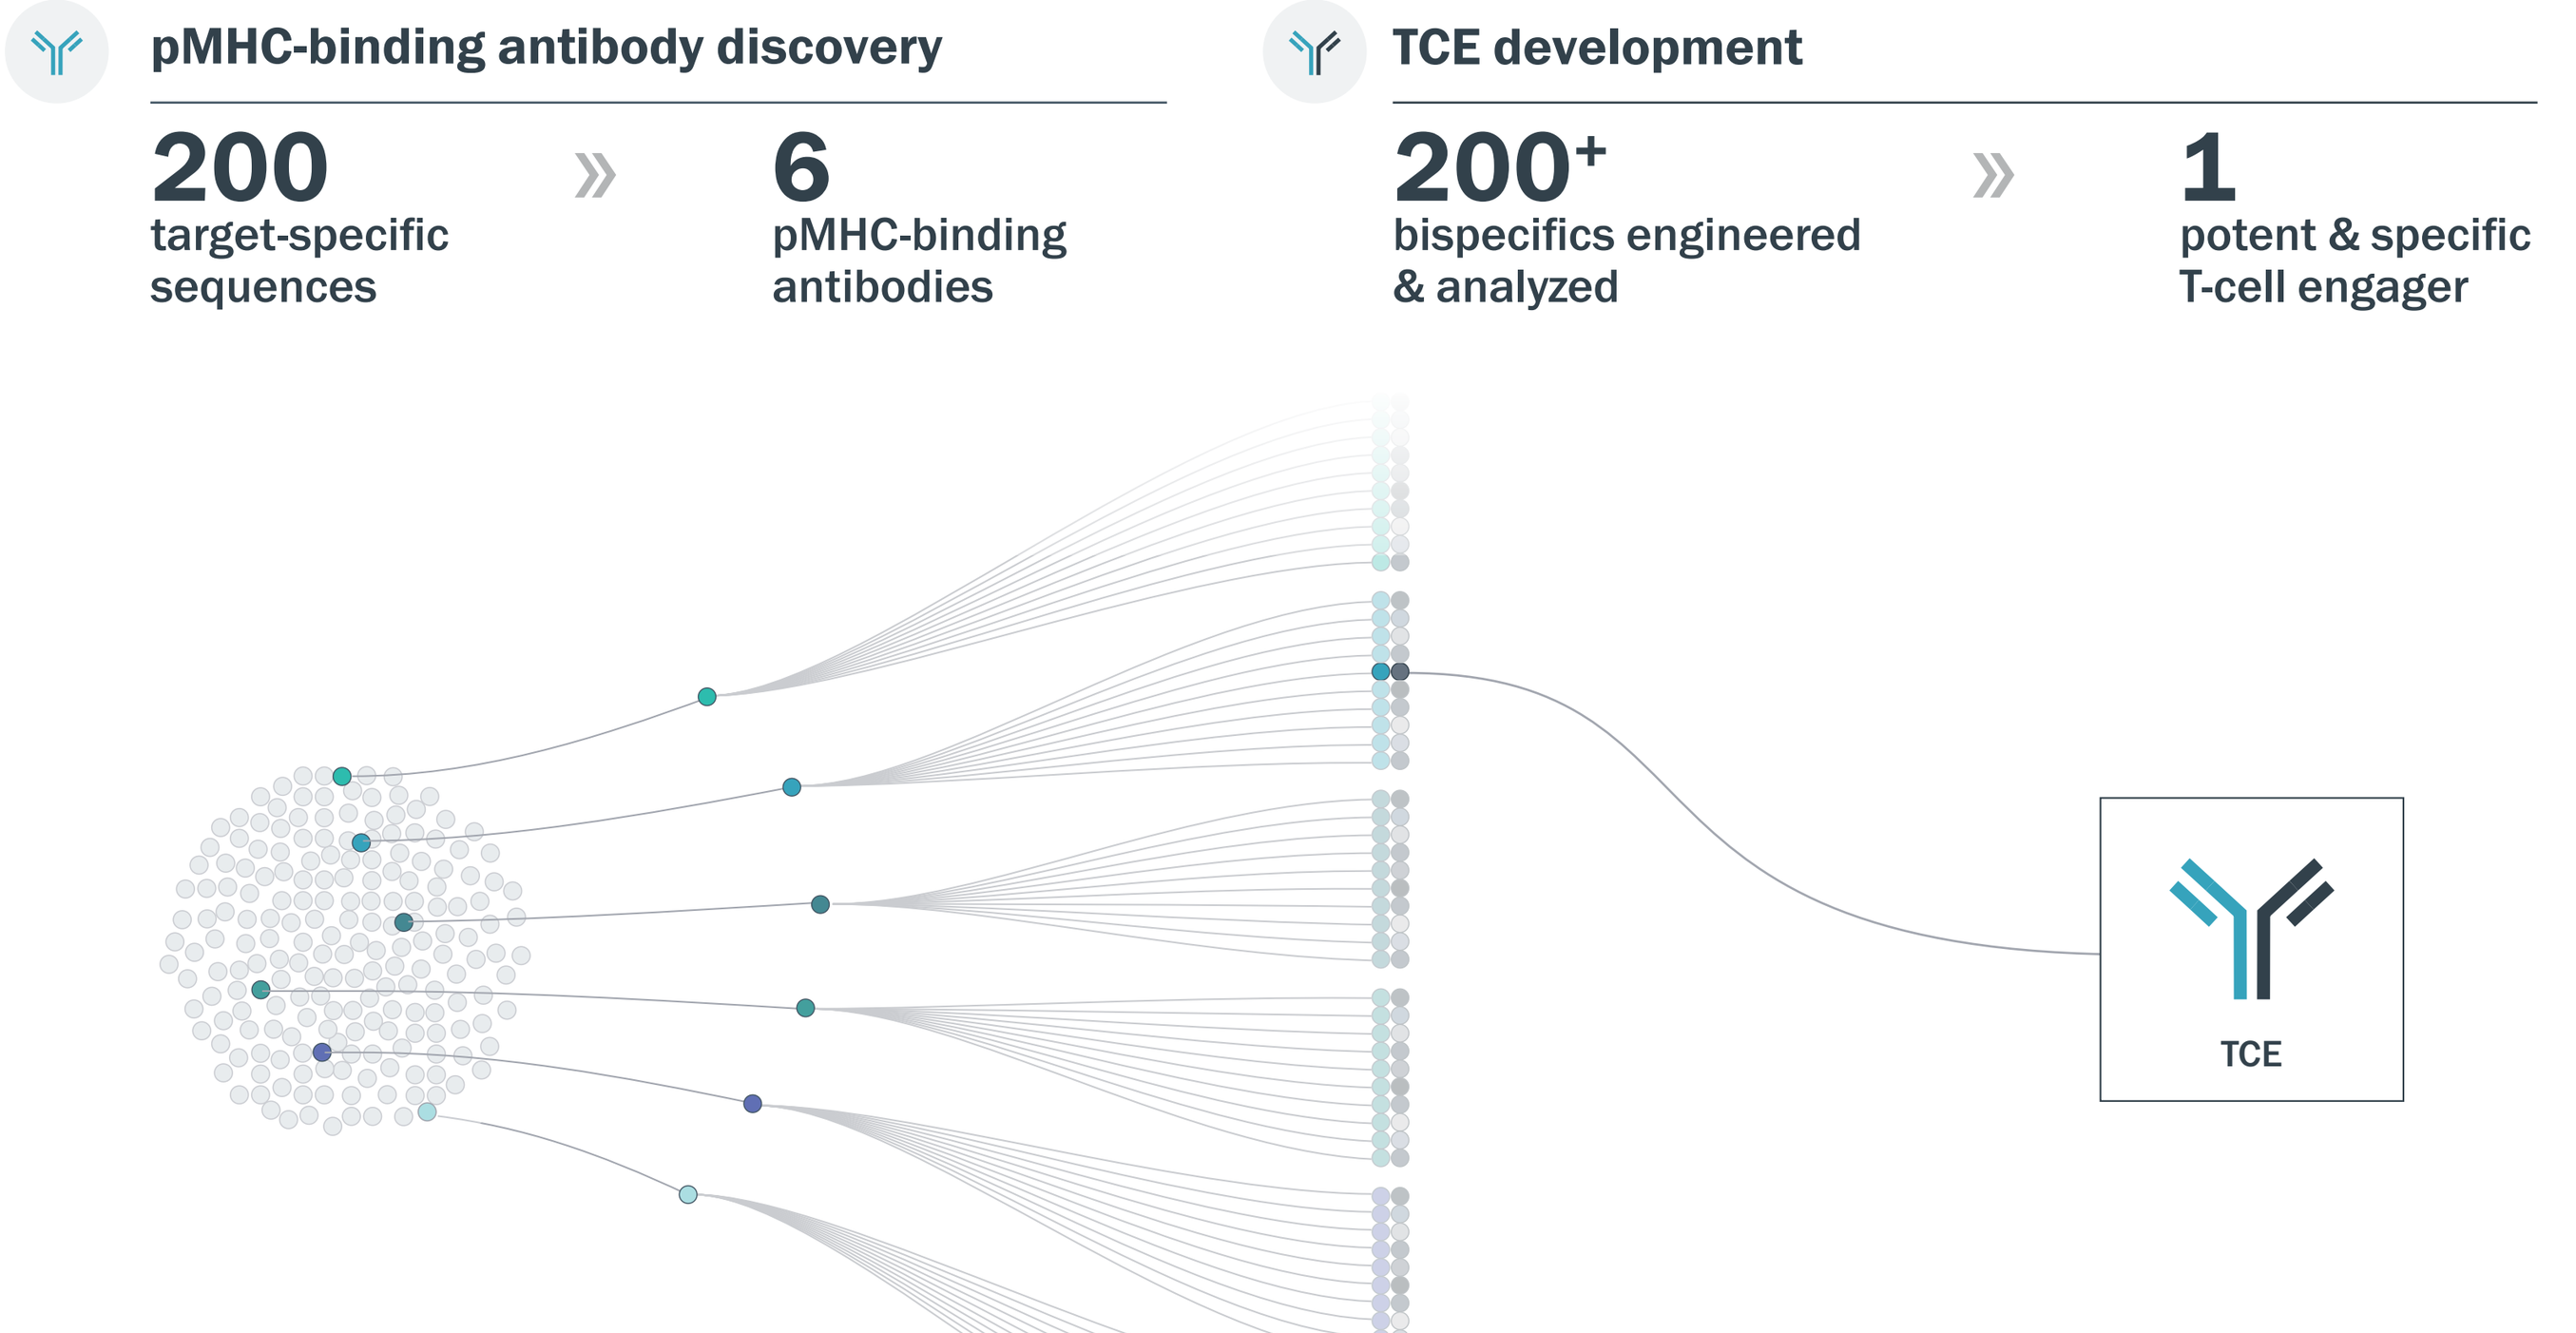

pMHC-binding antibody discovery
200
target-specific sequences
6
pMHC-binding antibodies
TCE development
200+
bispecifics engineered & analyzed
1
potent & specific T-cell engager
TCE

Figure 5. Deep screening of immune repertoires, high-throughput *in vitro* assessments, and a robust TCE platform enable identification of rare TCEs with high potency and specificity for pMHC targets. We used our proprietary single-cell screening platform to interrogate 1.5 million single cells. We identified 200 unique MAGE-A4-pMHC-binding antibody sequences, six of which had specificity and developability profiles suitable for TCE engineering. We leveraged our TCE platform, which includes novel CD3-binding antibodies that are differentiated from molecules commonly used for TCE development, to generate more than 200 1x1 bispecific TCEs. Following high-throughput functional and biophysical analyses, 12 were selected for in-depth assessment. Antibodies were produced at mid-scale for rigorous *in vitro* assessment, and one molecule with the potency and specificity required to target MAGE-A4-pMHC was identified for further preclinical assessment.

REFERENCES

1. Weekes MP, et al. (2010). *J Biomol Tech.* 21(3):108-115. PMID: 20808639.
2. Hong CW, et al. (2014). *FEBS Lett.* 588(2):350-355, doi: 10.1016/j.febslet.2013.10.025
3. Chandran SS, et al. (2019). *Immunol Rev.* 290(1):127-147, doi: 10.1111/imr.12772
4. Zhao Y, et al. (2007). *J. Immunol.* 179:5845–5854, doi: 10.4049/jimmunol.179.9.5845.
5. Border EC, et al. (2019). *OncoImmunol.* 8:e1532759, doi: 10.1080/2162402X.2018.1532759.
6. Foote J, Eisen HN. (1995). *Proc. Natl Acad. Sci. USA.* 92:1254–1256, doi: 10.1073/pnas.92.5.1254.
7. Rossjohn J, et al. (2015). *Annu. Rev. Immunol.* 33:169–200, doi: 10.1146/annurev-immunol-032414-112334.
8. Holland CJ, et al. (2020). *J. Clin. Invest.* 130:2673–2688, doi: 10.1172/JCI130562.
9. Schooten E, et al. (2018). *Cancer Treat Rev.* 67:54-62, doi: 10.1016/j.ctrv.2018.04.009.
10. de Puyraimond V, et al. (2023). *Journal for ImmunoTherapy of Cancer.* 11(Suppl 1):A1523, doi: 10.1136/jitc-2023-SITC2023.1367.
11. Weinzierl T, et al. (2021). (International Publication No. WO/2021/122875). World Intellectual Property Organization.
12. Fonseca AF, et al. (2023). *Font Immunol.* 14:1142573, doi: 10.3389/fimmu.2023.1142573.

COPYRIGHT © ABCELLERA

AbCellera's T-cell engager platform

We paired **six MAGE-A4-pMHC binders** with **19 antibodies from AbCellera's TCE platform** to generate and assess 200$^+$ bispecifics in a 1x1 format.

Learn more: Abstract 1868 | Section 22